Peripherally-Restricted Dual-Acting Kappa/Delta Opioid Agonist (CAV1001) Prevents Formalin-Induced Hyperalgesia

Craig T. Hartrick, MD, FIPP[1], Dominic Poulin, MSc[2], Allison Hartrick, MBA[1]
(1)Caventure Drug Discovery, (2)Charles River Laboratories, Montreal

Background

In the periphery, *kappa* (KOR) and *delta*-opioid (DOR) receptors are constitutively present. In an uninjured state peripheral *mu*-opioid receptors (MOR) play a minor role in the pain pathway; however, the MOR are responsible for opioid-induced constipation. The DOR become active following the induction of the inflammatory response.[1]

Objective

The relative contribution of DOR agonism following the induction of inflammation was examined comparing a novel dual-acting peripherally -restricted DOR/KOR agonist (CAV1001) to a pure KOR peripherally-restricted agonist (ICI204448) in the formalin model.

- Plantar injection (*sc*) of formalin causes a bi-phasic nocifensive behavioral response;
- The early phase (phase 1: acute injury) is followed by an interphase without discernible nociceptive reactions, after which the late phase ensues (phase 2: inflammatory response);
- Phase 2 is used as a pharmacodynamic surrogate of central sensitization.

Methods

Following IACUC approval, mice were randomly pretreated with inert vehicle, ICI204448 1 mg/kg, ICI204448 10 mg/kg, CAV1001 1 mg/kg, or CAV1001 10 mg/kg; Spontaneous nocifensive behaviors were blindly assessed (video recording).

Study Design

Groups	Group Treatment	Dose Level (mg/kg)	Route	Dose Vol. (mL/kg)	Pre-Treatment Time	N
1	Vehicle (1:1:8 Ethanol: Tween 80: 0.9% Saline)	0	IP	20	30 min	8
2	ICI204448 – Low Dose	1	IP	20	30 min	8
3	ICI204448 – High Dose	10	IP	20	30 min	8
4	CAV1001 – Low Dose	1	IP	20	30 min	8
5	CAV1001 – High Dose	10	IP	20	30 min	8

Results

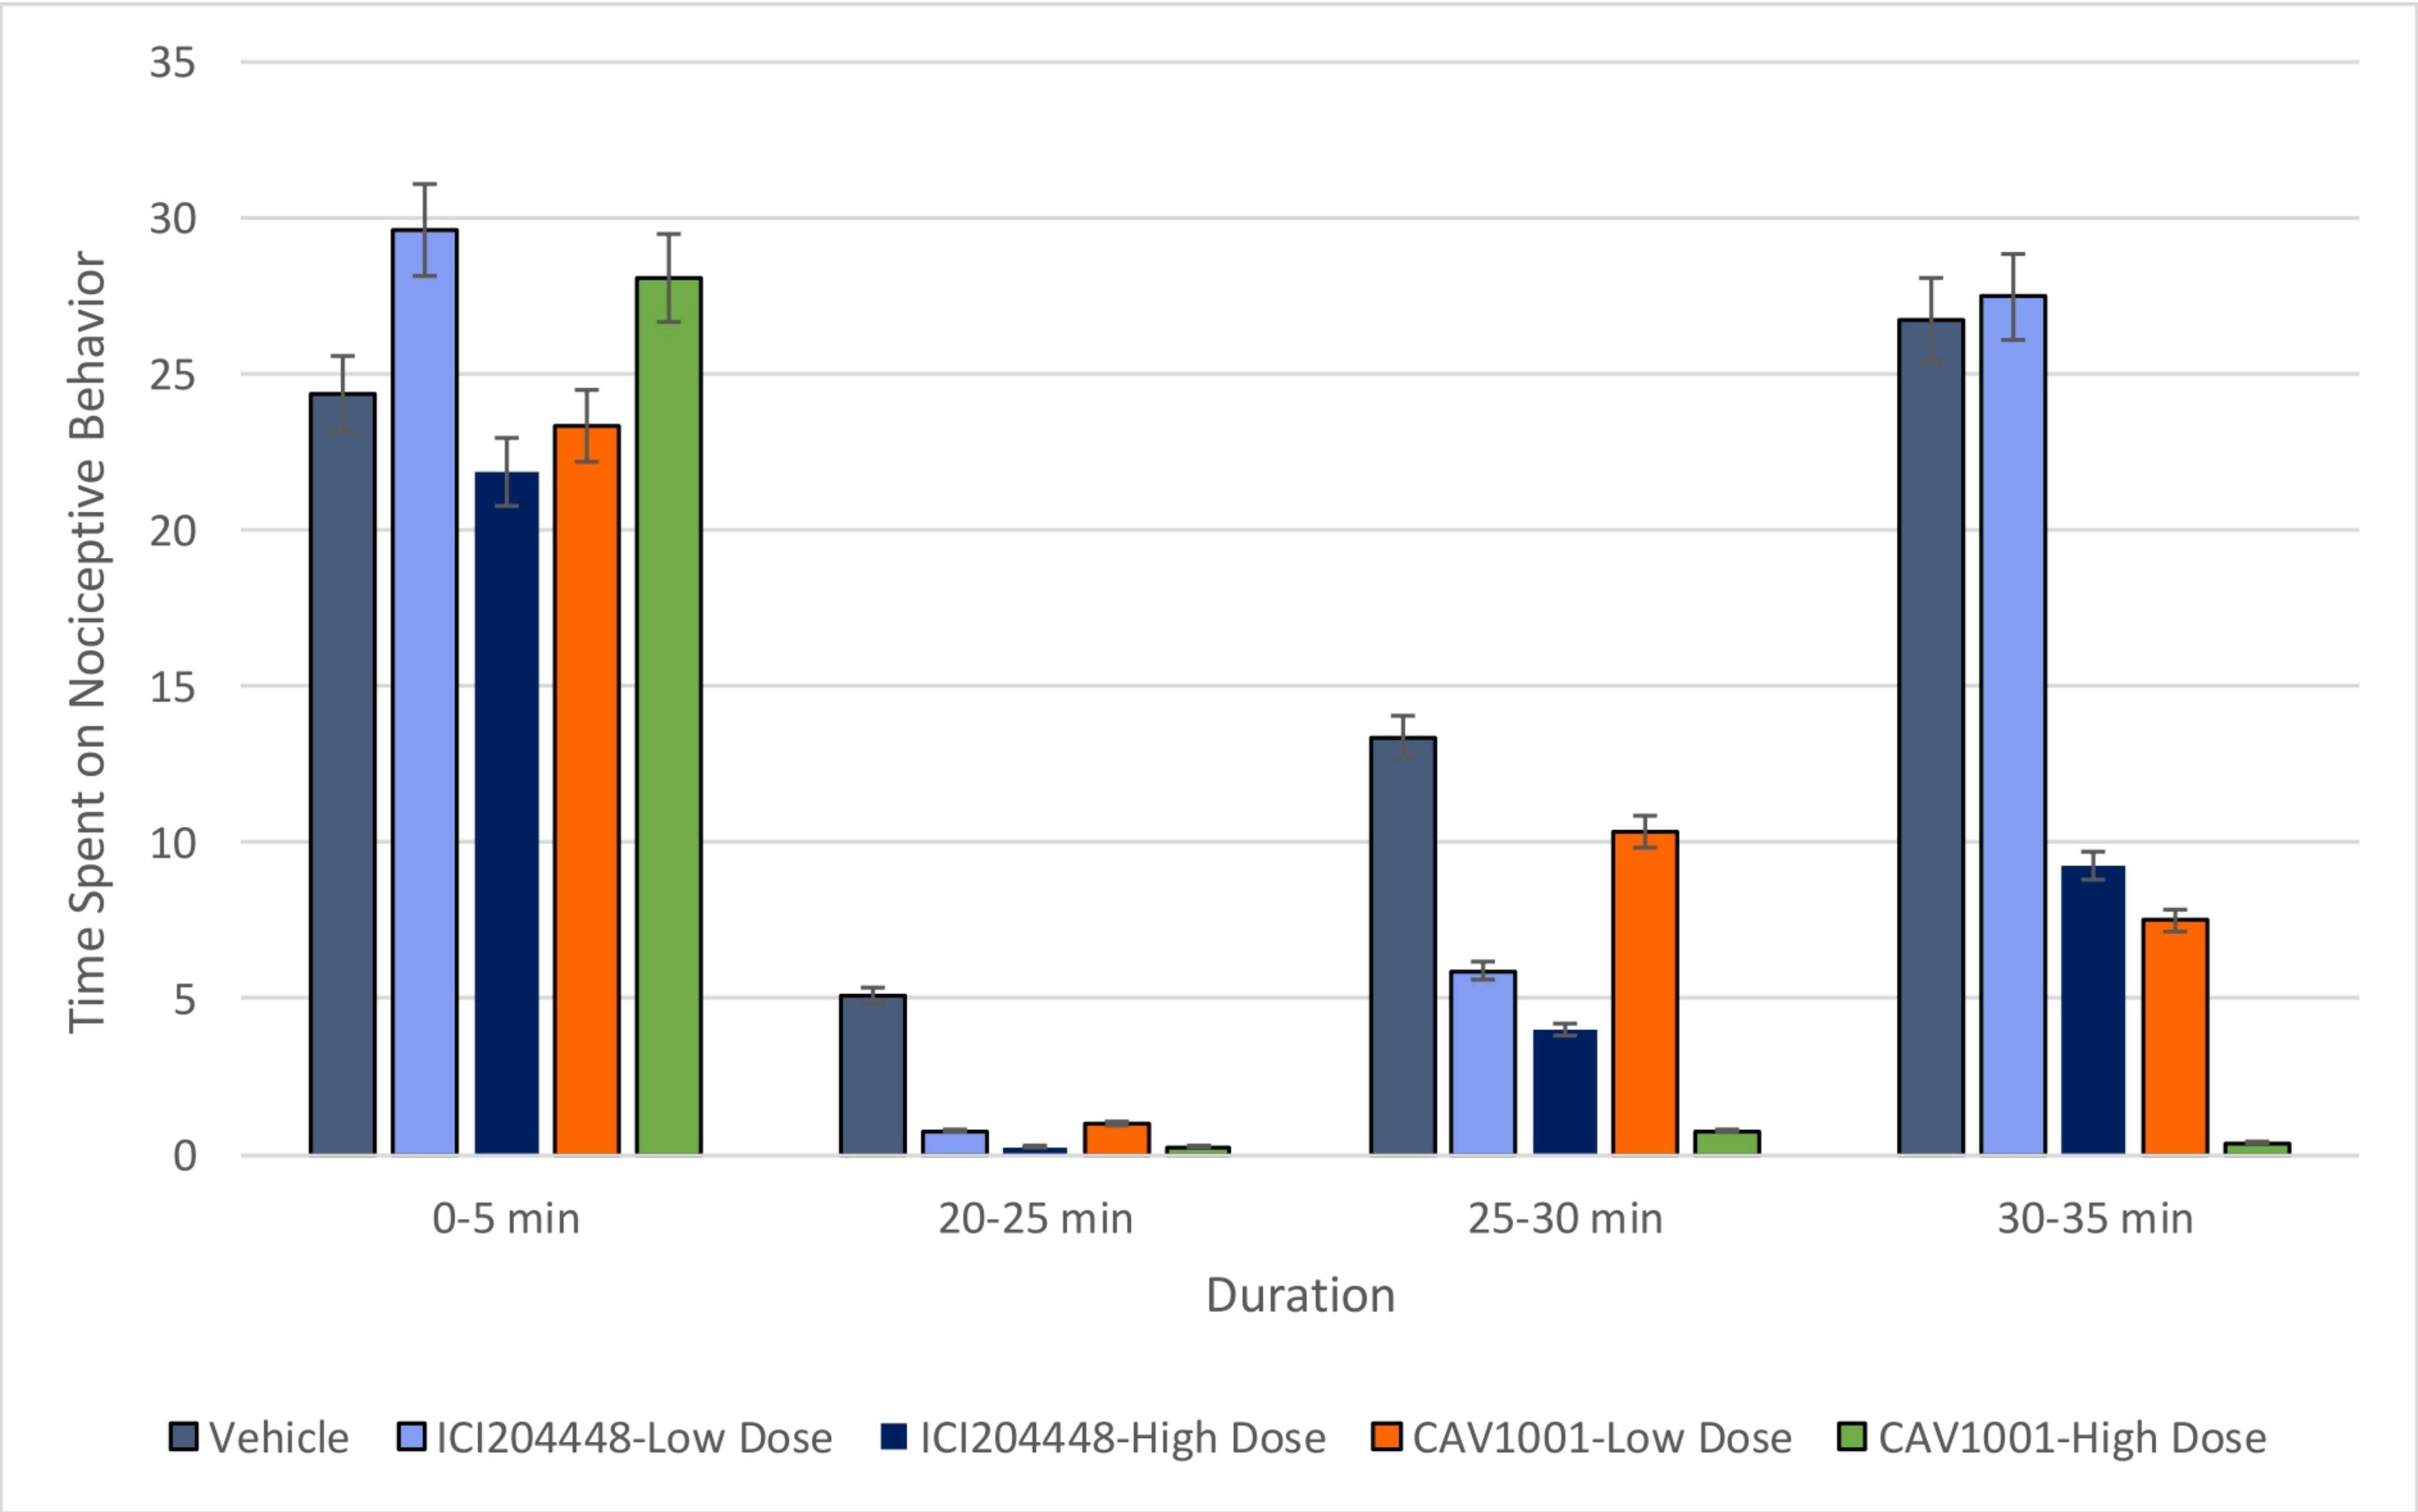
35
30
25
20
15
10
5
0
Time Spent on Nociceptive Behavior
0-5 min
20-25 min
25-30 min
30-35 min
Duration
Vehicle
ICI204448-Low Dose
ICI204448-High Dose
CAV1001-Low Dose
CAV1001-High Dose

Comparison to Peripherally-Restricted KOR Agonist

Comparison	Timeframe	pValue
CAV1001 (High Dose) v. Vehicle	30-35 min	p<0.03
CAV1001 (High Dose) v. Vehicle	20-35 min	p<0.003
CAV1001 (High Dose) v. KOR Agonist (High Dose)	20-35 min	p<0.01
KOR Agonist (High Dose) v. Vehicle	20-35 min	p<0.05

- Neither agent was effective in reducing the acute (0-5 minutes) response to formalin injection (phase 1);
- CAV1001 1 mg/kg was as effective as ICI204448 10 mg/kg in reducing formalin-induced responses at 20-35 minutes;
- CAV1001 10 mg/kg was significantly more effective than ICI204448 10 mg/kg (phase 2; $p<0.01$);
- Moreover, CAV1001 10 mg/kg effectively prevented the development of the phase 2 hyperalgesic response ($p<0.003$).

Conclusion

The apparent synergistic effect of simultaneous *kappa* and *delta* opioid agonism in the presence of inflammation suggests potential advantage over pure peripherally-restricted *kappa* agonists in inflammatory pain settings and further supports a preemptive effect that can eliminate the hyperalgesic response in painful inflammatory states.

Reference

[1]Brackley AD, Gomez R, Akopian AN, Henry MA, Jeske NA. Cell Rep. 2016; 16(10): 2686–2698.

Contact: Dr. Craig T Hartrick | chartrick@caventuredrugdiscovery.com or Allison Hartrick | ahartrick@caventuredrugdiscovery.com

Caventure Drug Discovery