The Efficacy of a Dual-Acting, Peripherally-Restricted kappa/delta Opioid Agonist (CAV1001) in Neuropathic Pain in the Rat

Craig T. Hartrick, MD, FIPP[1], Thomas Van Valkenburg, BA[2], Jim Pomonis, PhD[2] , Allison Hartrick, MBA[1]
(1)Caventure Drug Discovery, (2)American Preclinical Services

Background

Nerve injury can precipitate a neuroimmune inflammatory response.[1] This response might lead to the activation of peripheral delta opioid receptors, allowing *delta* opioid agonists in the periphery to become analgesic directly and through allosteric modulation of peripheral *kappa* opioid receptors.[2]

Purpose

This study evaluated the efficacy of a single intraperitoneal injection of CA1001 (a novel peripherally-restricted dual-acting *kappa/delta*-opioid agonist) and the comparator, gabapentin, in the spinal nerve ligation (SNL) model for neuropathic pain in the rat.

Methods

- Following IACUC approval, neuropathy was induced by surgically ligating the 5th and 6th lumbar spinal nerves (L5 and L6);
- Mechanical sensitivity was assessed via paw compression thresholds (PCTs) using a digital Randall-Selitto device;
- 50 animals that met the inclusion criteria were randomly assigned to 5 groups with 10 animals per group (Power: 80%);
- Animals were administered a single dose of vehicle, CAV1001 (1 mg/kg, 5 mg/kg, or 10 mg/kg IP), or control compound (gabapentin 100 mg/kg PO; active control: internal validity) on day 0 (15 days after SNL) and PCTs were determined 1, 2, and 4 hours after compound administration;
- All behavioral evaluations were performed by a blinded observer.

Study Design: Neuropathic Pain (SNL)

Test System ID: Species: Breed: Sex			Rat: Sprague-Dawley: Male			
Group #	Treatment	N	Dose (mg/kg)	Vol. (mL/kg)	Route	Day of Admin/ Frequency
1	Vehicle (Ethanol: Tween 80: Normal Saline – 1:1:8)	10	N/A	5	IP	Day 0 / 1x
2	CAV1001	10	1	5	IP	Day 0 / 1x
3	CAV1001	10	5	5	IP	Day 0 / 1x
4	CAV1001	10	10	5	IP	Day 0 / 1x
5	Gabapentin	10	100	5	IP	Day 0 / 1x

Results

Neuropathic Pain: Hyperalgesia Development

Unpaired t-test, two-tailed, Ipsilateral vs. Contralateral			
Time Point	t	Df	p-Value
Pre-Injury Baseline	0.6803	18	0.505
Pre-Dosing Baseline	4.59	18	0.0002
1 Hour	6.542	18	<0.0001
2 Hour	4.098	18	0.0007
4 Hour	7.304	18	<0.0001

Gabapentin significantly reversed SNL-induced mechanical hyperalgesia at 1, 2, and 4-hours (p<0.01 versus vehicle, t test).

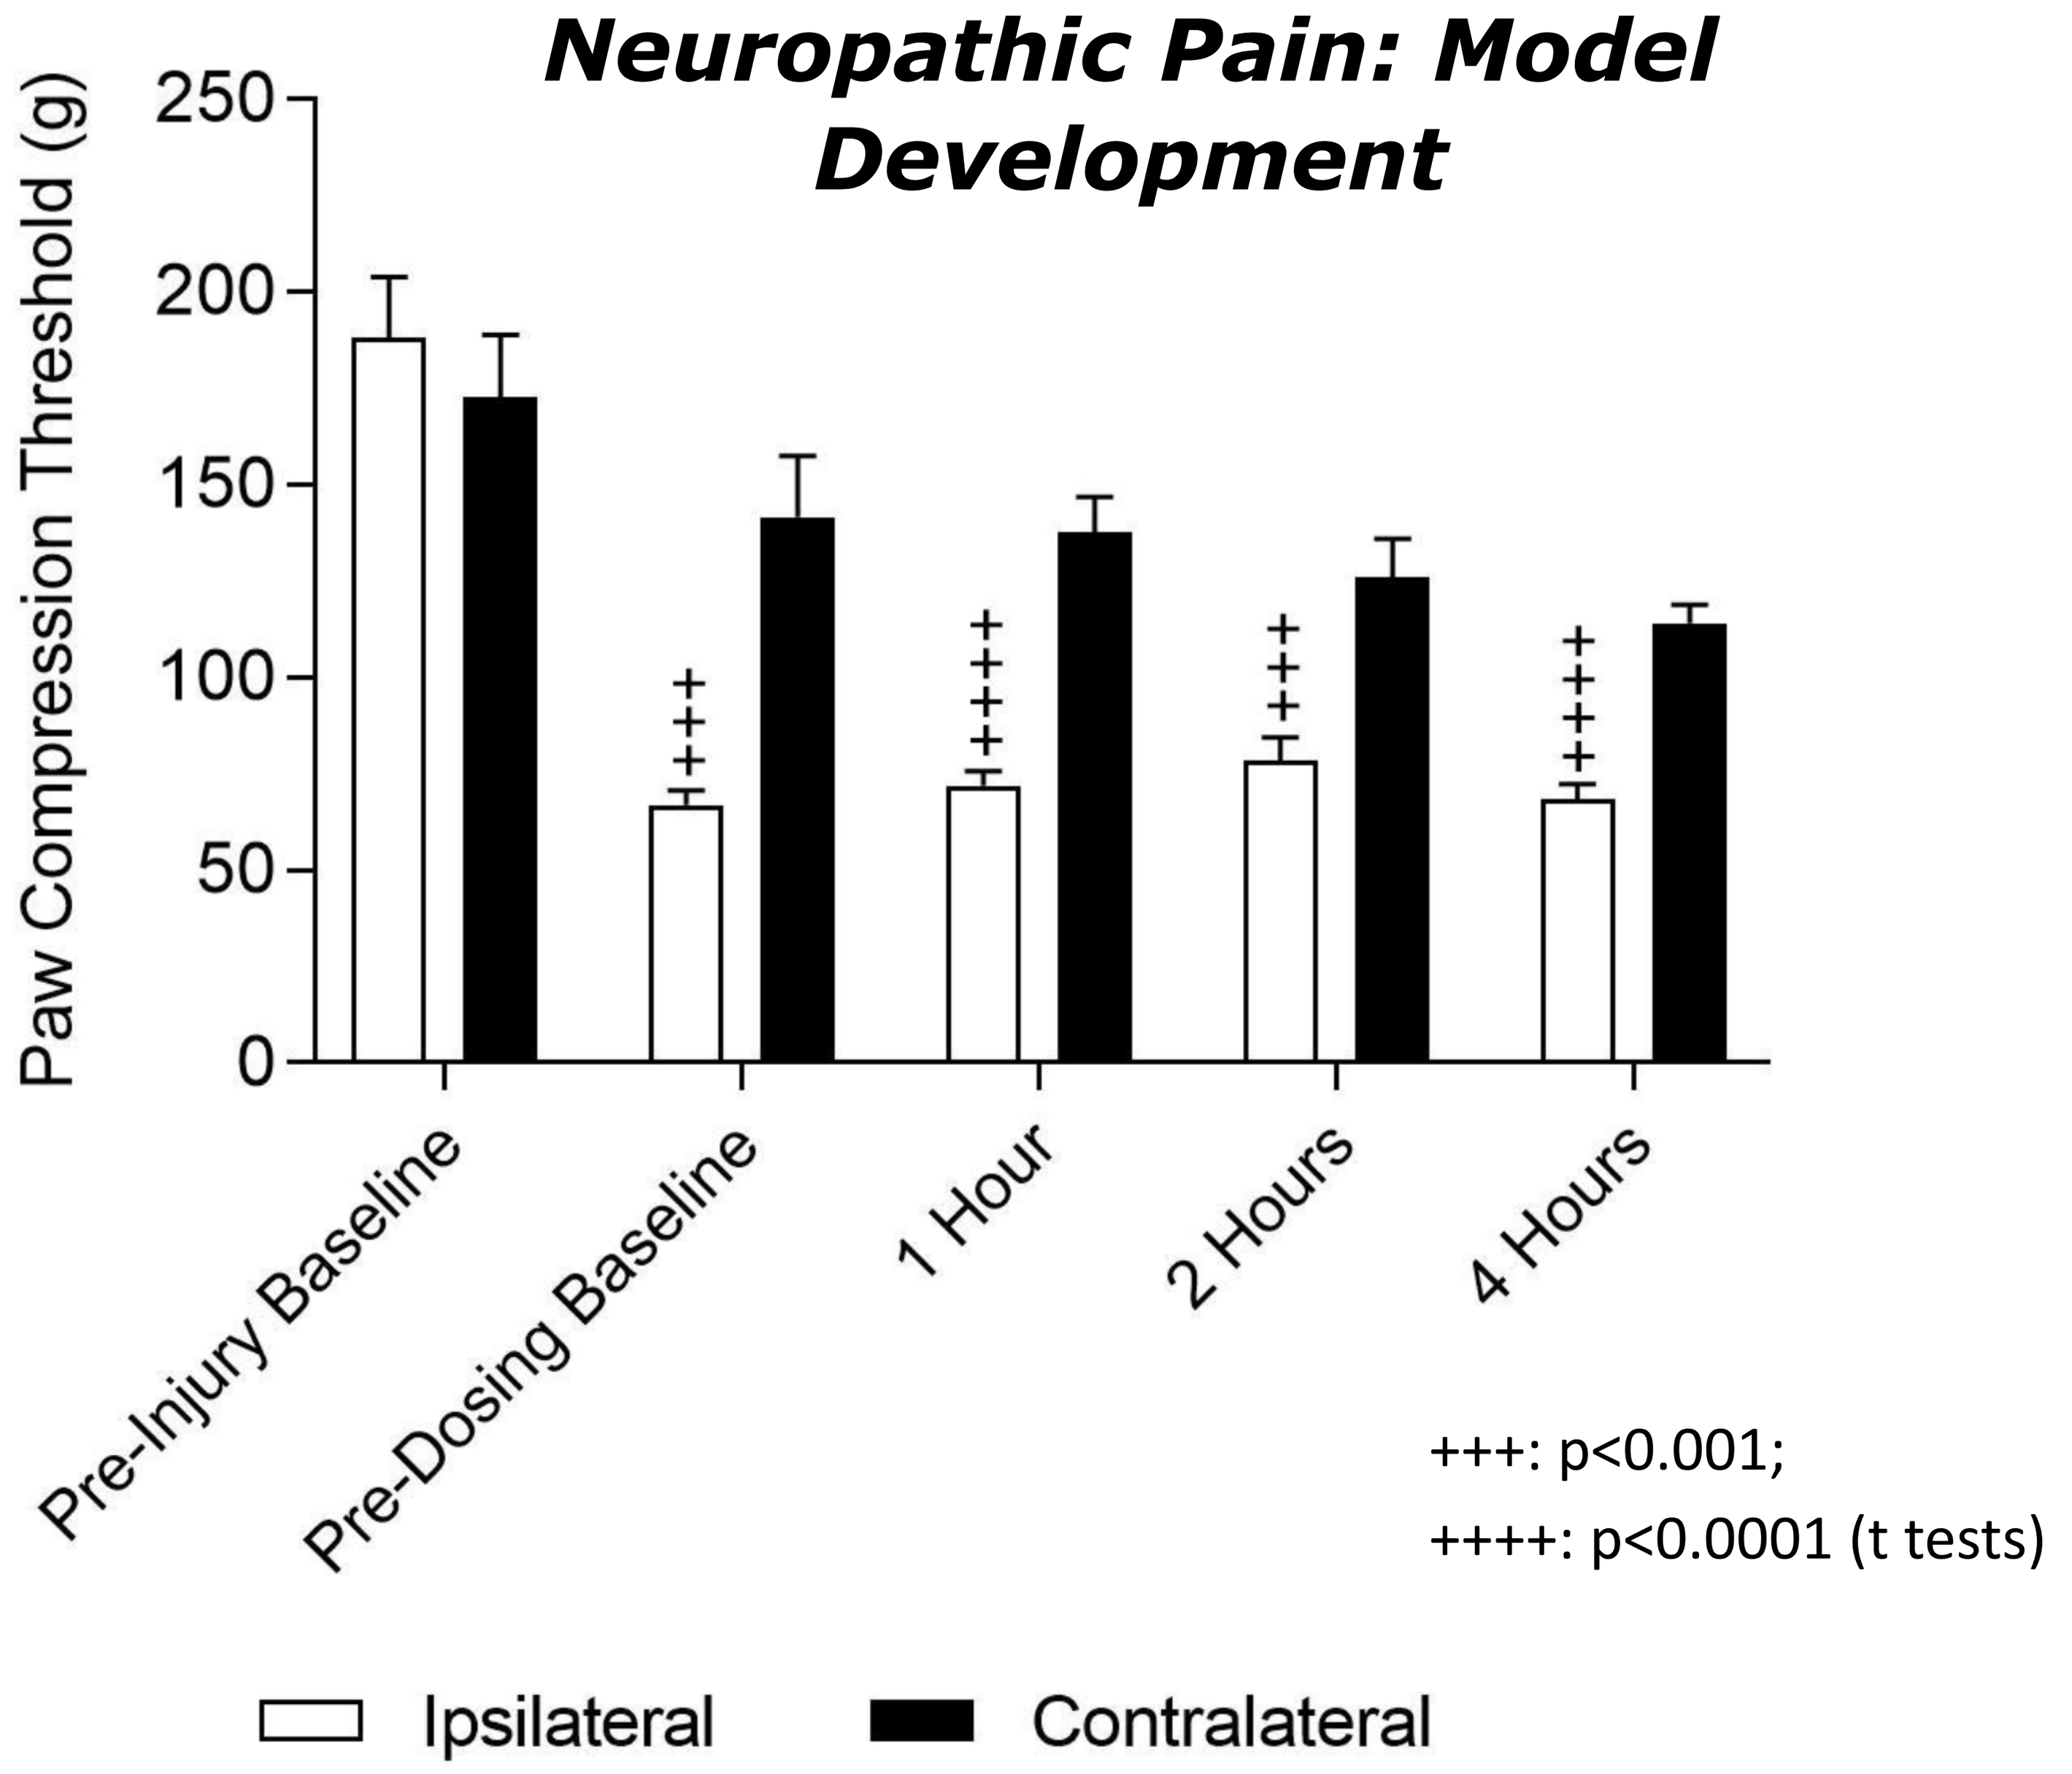
Neuropathic Pain: Model Development
Paw Compression Threshold (g)
250
200
150
100
50
0
Pre-Injury Baseline
Pre-Dosing Baseline
1 Hour
2 Hours
4 Hours
+++: p<0.001;
++++: p<0.0001 (t tests)
Ipsilateral
Contralateral

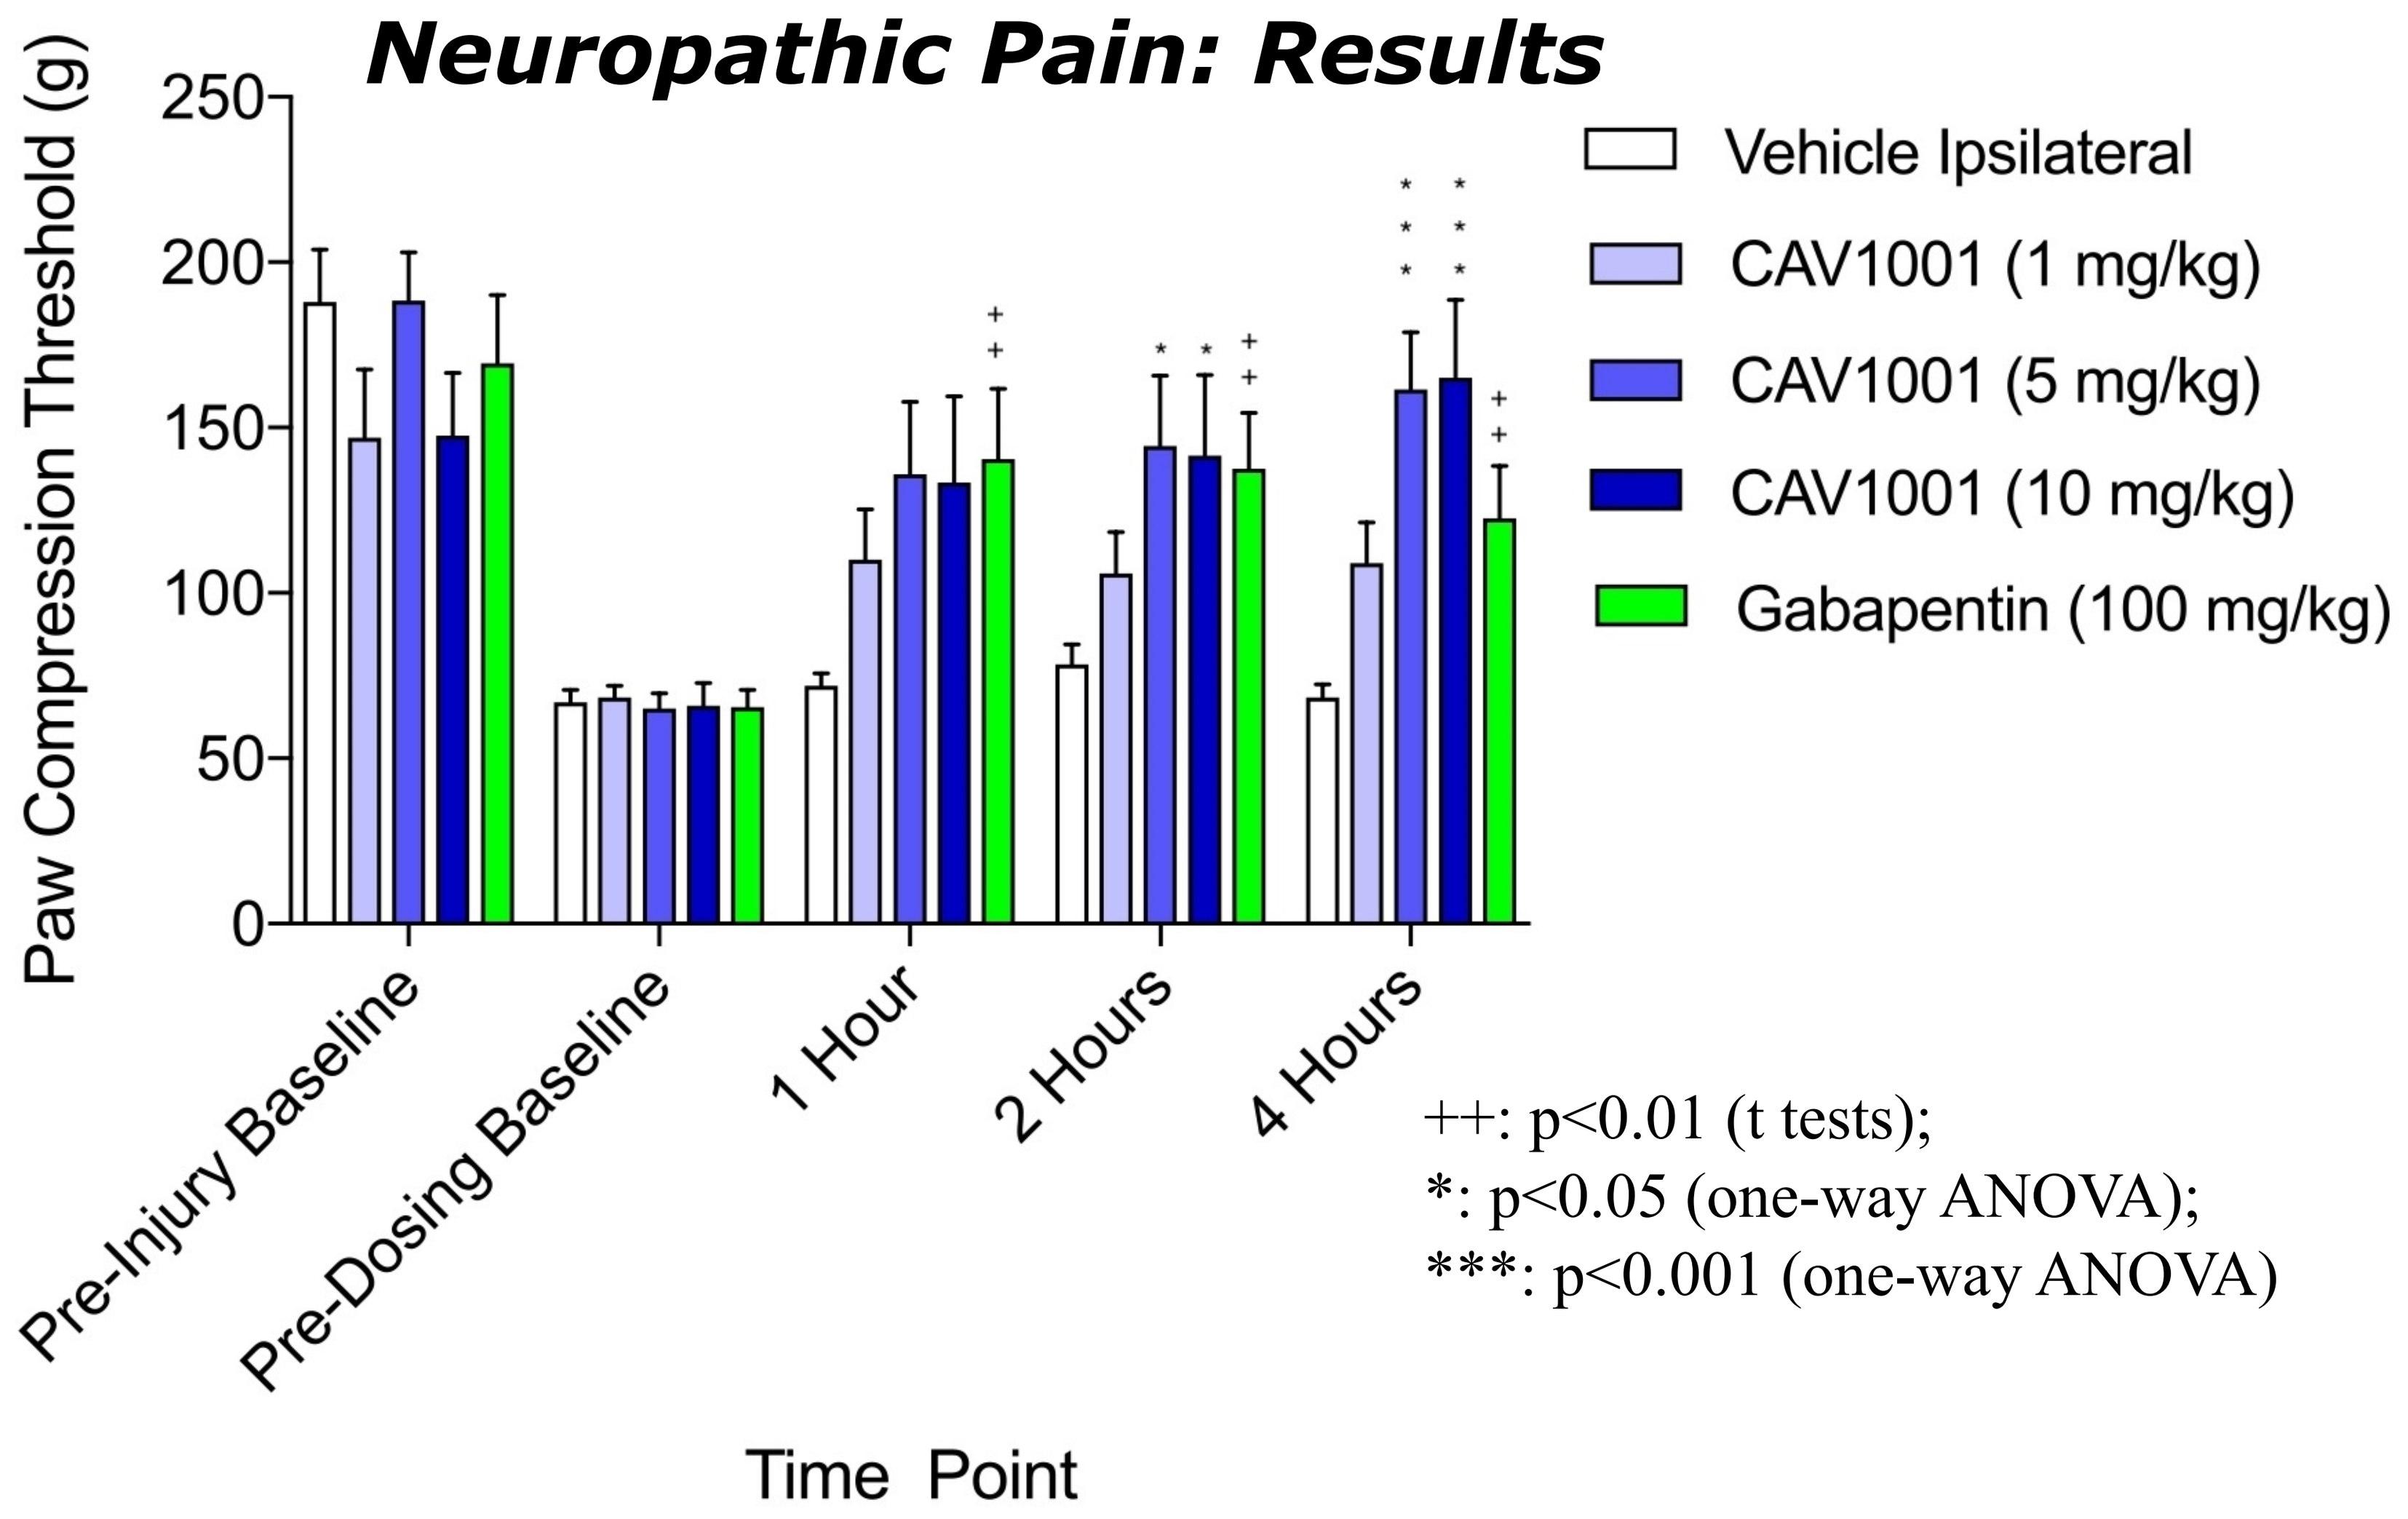
Neuropathic Pain: Results
Paw Compression Threshold (g)
250
200
150
100
50
0
Pre-Injury Baseline
Pre-Dosing Baseline
1 Hour
2 Hours
4 Hours
Time Point
Vehicle Ipsilateral
CAV1001 (1 mg/kg)
CAV1001 (5 mg/kg)
CAV1001 (10 mg/kg)
Gabapentin (100 mg/kg)
++: p<0.01 (t tests);
*: p<0.05 (one-way ANOVA);
***: p<0.001 (one-way ANOVA)

- CAV1001 at the 1 mg/kg dose did not significantly reverse mechanical hyperalgesia at any of the time points tested;
- The 5 mg/kg and 10 mg/kg doses of CAV1001 did not significantly reverse mechanical hyperalgesia at the 1-hour post-dosing time point but did significantly reverse mechanical hyperalgesia at the 2-hour (p<0.05 versus vehicle by one-way ANOVA) and 4-hour (p<0.001 versus vehicle by one-way ANOVA) post-dosing time points.

Conclusion

The reversal of mechanical hyperalgesia by CAV1001 at 5 mg/kg and 10 mg/kg did not differ significantly from the active control, gabapentin.

References

[1]Totsch SK, Sorge RE. Immune system involvement in specific pain conditions. Molecular Pain. 2017;13:1-17.
[2]Brackley AD, Gomez R, Akopian AN, Henry MA, Jeske NA. Cell Rep. 2016; 16(10): 2686–2698.

Contact: Dr. Craig T Hartrick | chartrick@caventuredrugdiscovery.com or Allison Hartrick | ahartrick@caventuredrugdiscovery.com

Caventure Drug Discovery