MOSPD2 regulates the activation state of αLβ2 integrin to control monocyte migration

Itzhak Mendel[1], Yaniv Salem[1], Oshrat Propheta-Meiran[1], Pinhas Kafri[1], Niva Yacov[1] and Eyal Breitbart[1]

[1]VBL Therapeutics

VBL THERAPEUTICS

Abstract

Monocytes are innate immune cells that drive the chronicity of various inflammatory diseases, making them an attractive target for therapeutic intervention. Monocyte migration to inflamed tissues involves multiple steps of interaction with the vascular endothelium and the extra-cellular matrix (ECM), a process mainly mediated through conformational transitions in cell surface integrins. We previously described Motile sperm domain-containing protein 2 (MOSPD2) as a surface protein expressed on myeloid cells that is essential for the migration of monocytes and a key regulator of inflammatory disease pathogenesis. Therefore, a possible role for MOSPD2 in regulating adhesion and integrin activation in monocytes was assessed.

Silencing of MOSPD2 expression in THP-1 monocytic cell line significantly increased the adhesion of monocytes to various ECM ligands of integrins. Employing VB-601, a humanized anti-human MOSDP2 monoclonal antibody, on wild-type THP-1 cells or primary human CD14 monocytes similarly increased cell adhesion to ECM ligands as well as to adhesion molecules. At the molecular level, silencing of MOSPD2 or blocking MOSPD2 using VB-601 led to a transition in integrin αLβ2 conformation into active high affinity binding-form and to the phosphorylation of adhesion-associated pathways. Co-precipitation experiments show that MOSPD2 binds integrin-β2, but not integrin-β1.

Our results reveal a novel regulatory mechanism on monocyte migration in which MOSPD2 governs the balance between monocyte adhesion and release from adhesion molecules and ECM. Targeting MOSPD2 has the potential to treat inflammatory diseases by inhibiting monocyte infiltration into inflamed tissues.

Introduction

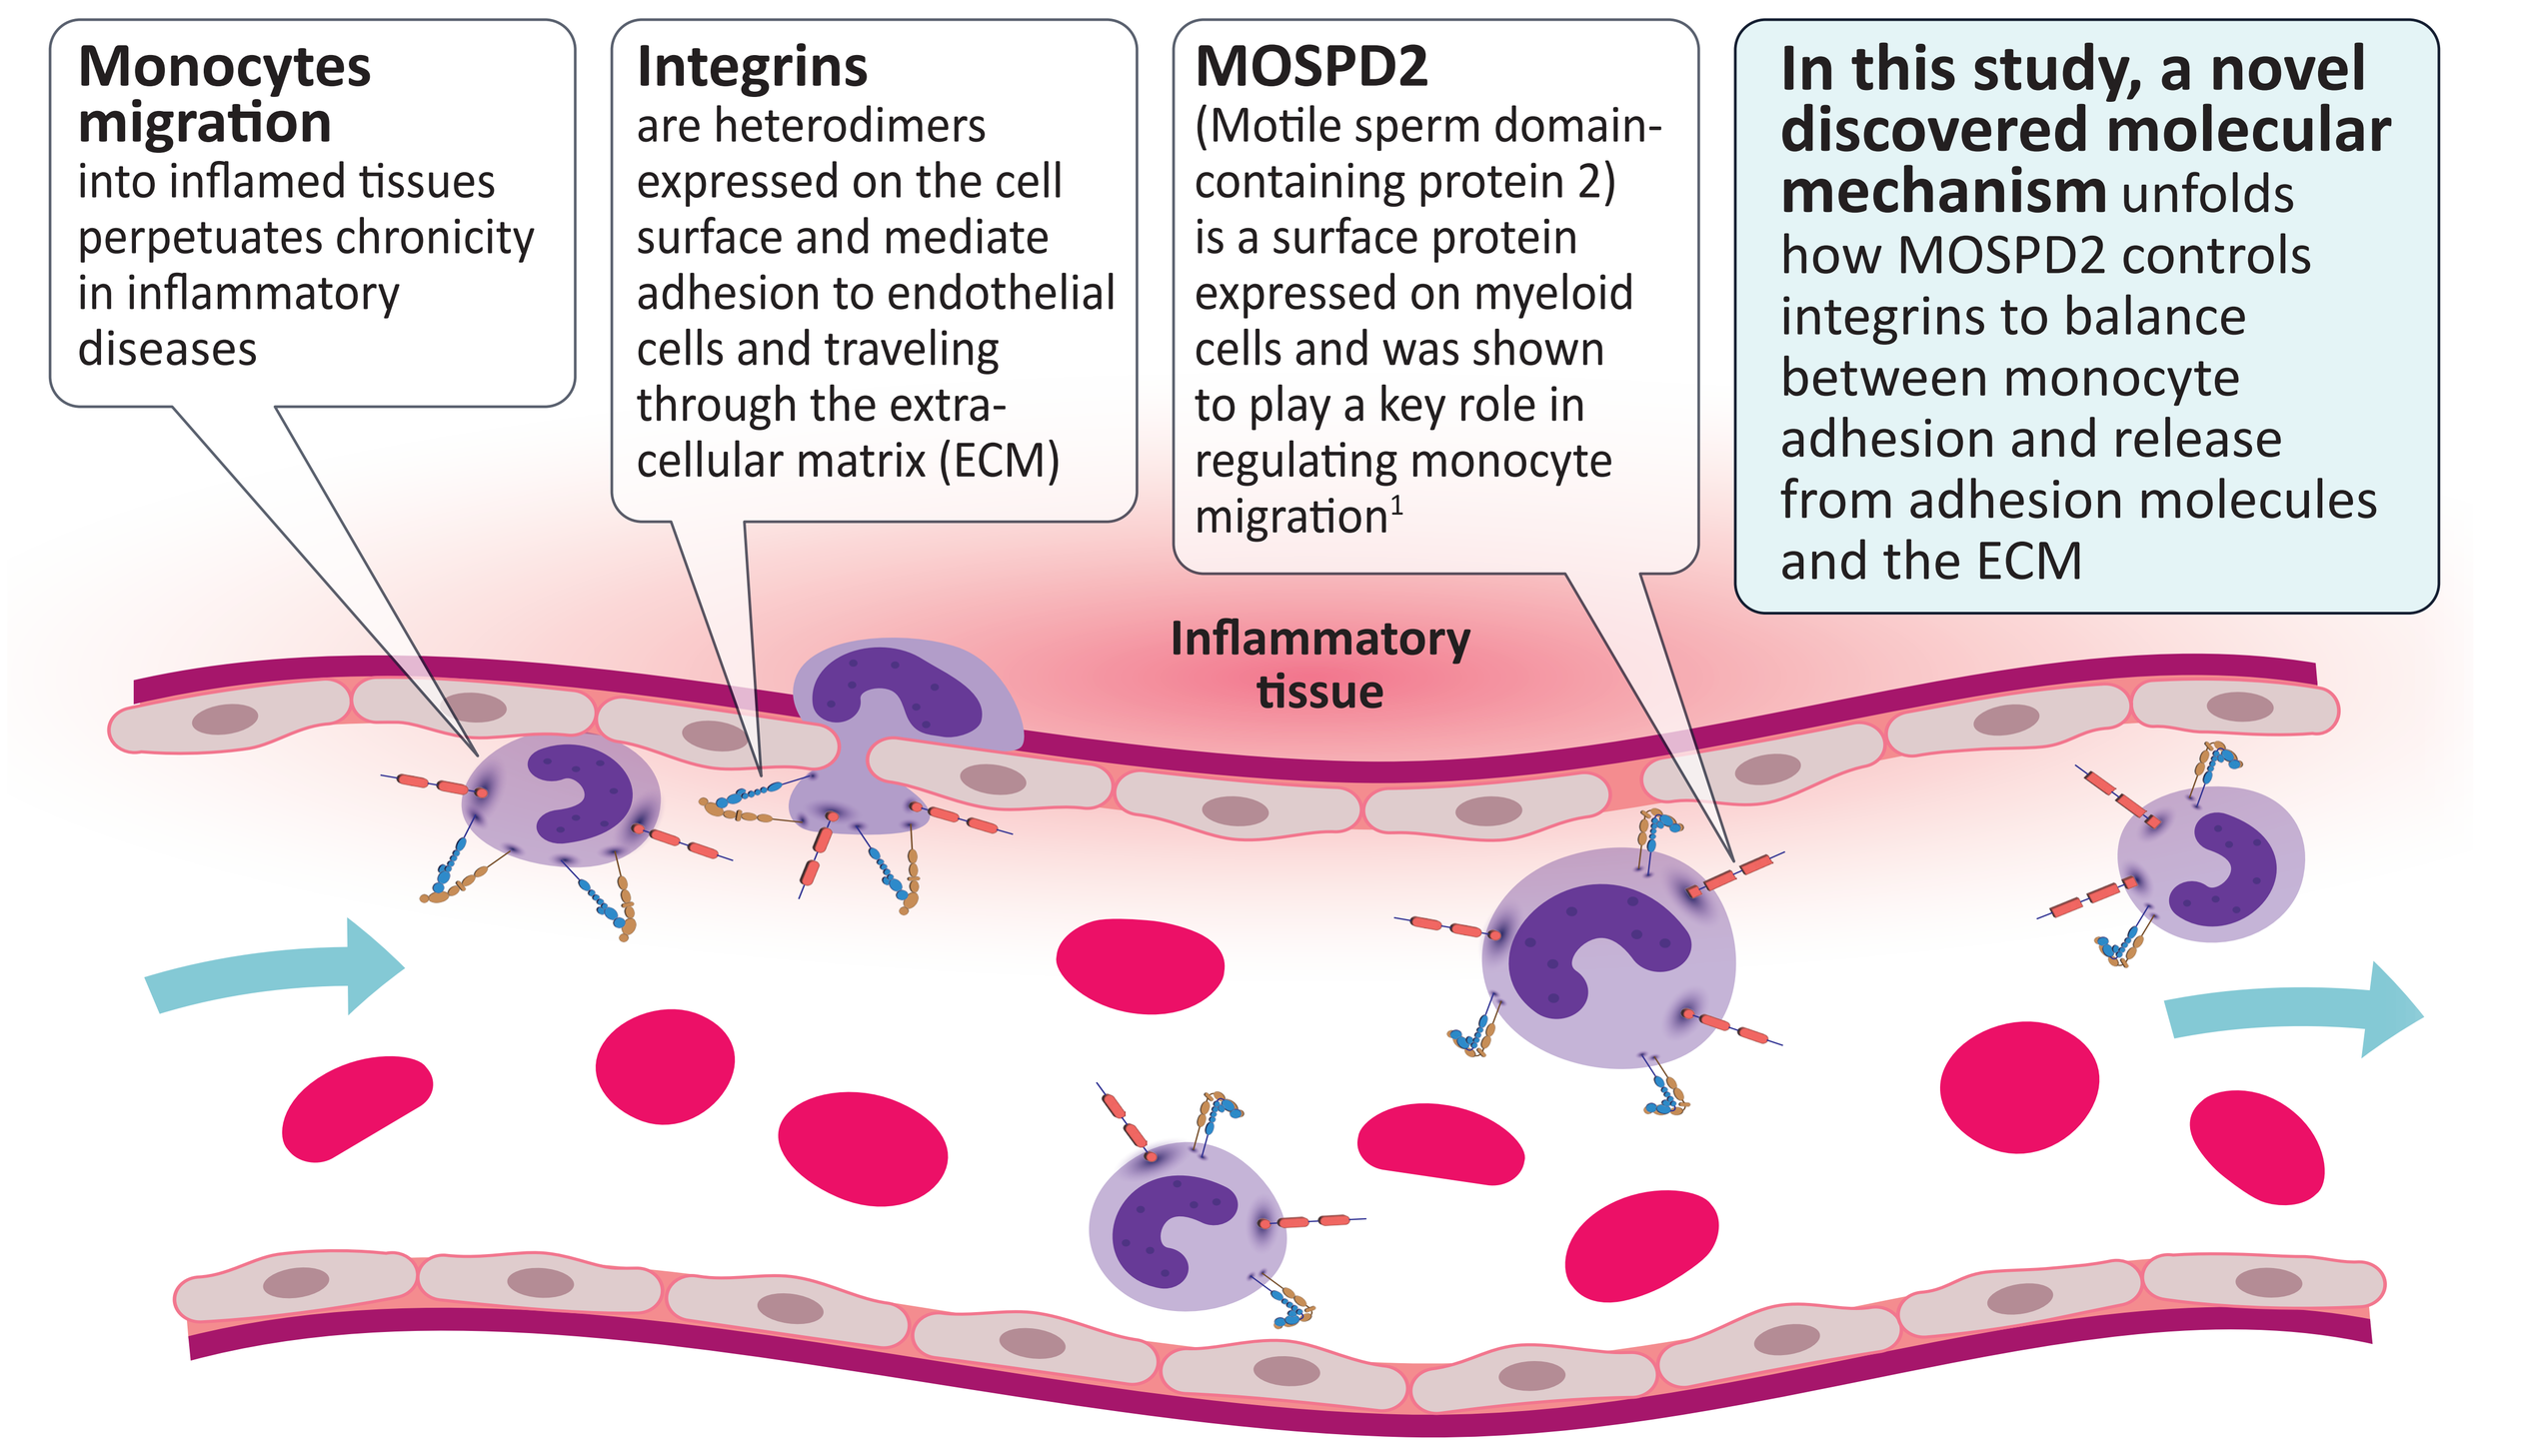
Monocytes migration into inflamed tissues perpetuates chronicity in inflammatory diseases
Integrins are heterodimers expressed on the cell surface and mediate adhesion to endothelial cells and traveling through the extra-cellular matrix (ECM)
MOSPD2 (Motile sperm domain-containing protein 2) is a surface protein expressed on myeloid cells and was shown to play a key role in regulating monocyte migration[1]
In this study, a novel discovered molecular mechanism unfolds how MOSPD2 controls integrins to balance between monocyte adhesion and release from adhesion molecules and the ECM
Inflammatory tissue

- MOSPD2-deficient mice were protected from the development of CNS inflammation in the EAE model of multiple sclerosis[2]
- Treatment with a mouse anti-MOSPD2 mAb inhibited disease progression in autoimmune inflammatory models for multiple sclerosis, rheumatoid arthritis and colitis
- We have developed VB-601, a humanized mAb that targets human MOSPD2, as a potential therapy for the treatment of autoimmune diseases

Results

Silencing MOSPD2 increases adhesion of monocytes to ECM molecules (THP-1 cell line)

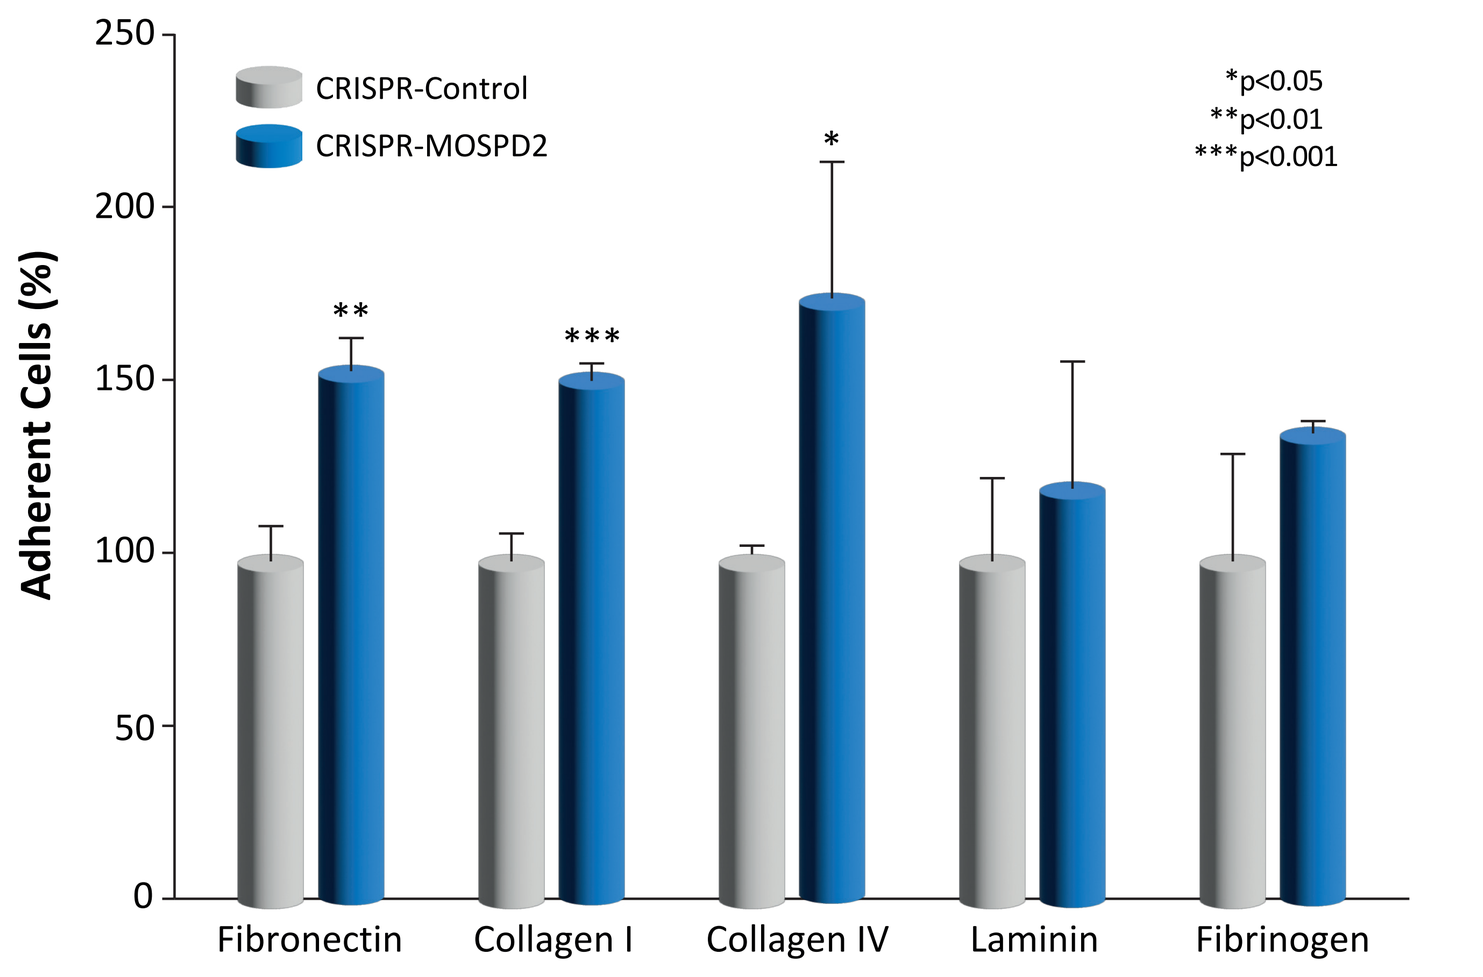
250
200
150
100
50
0
Adherent Cells (%)
CRISPR-Control
CRISPR-MOSPD2
*p<0.05
**p<0.01
***p<0.001
**

*
Fibronectin
Collagen I
Collagen IV
Laminin
Fibrinogen

Anti-MOSPD2 mAb VB-601 inhibits migration and increases adhesion of human primary monocytes in a dose-dependent manner

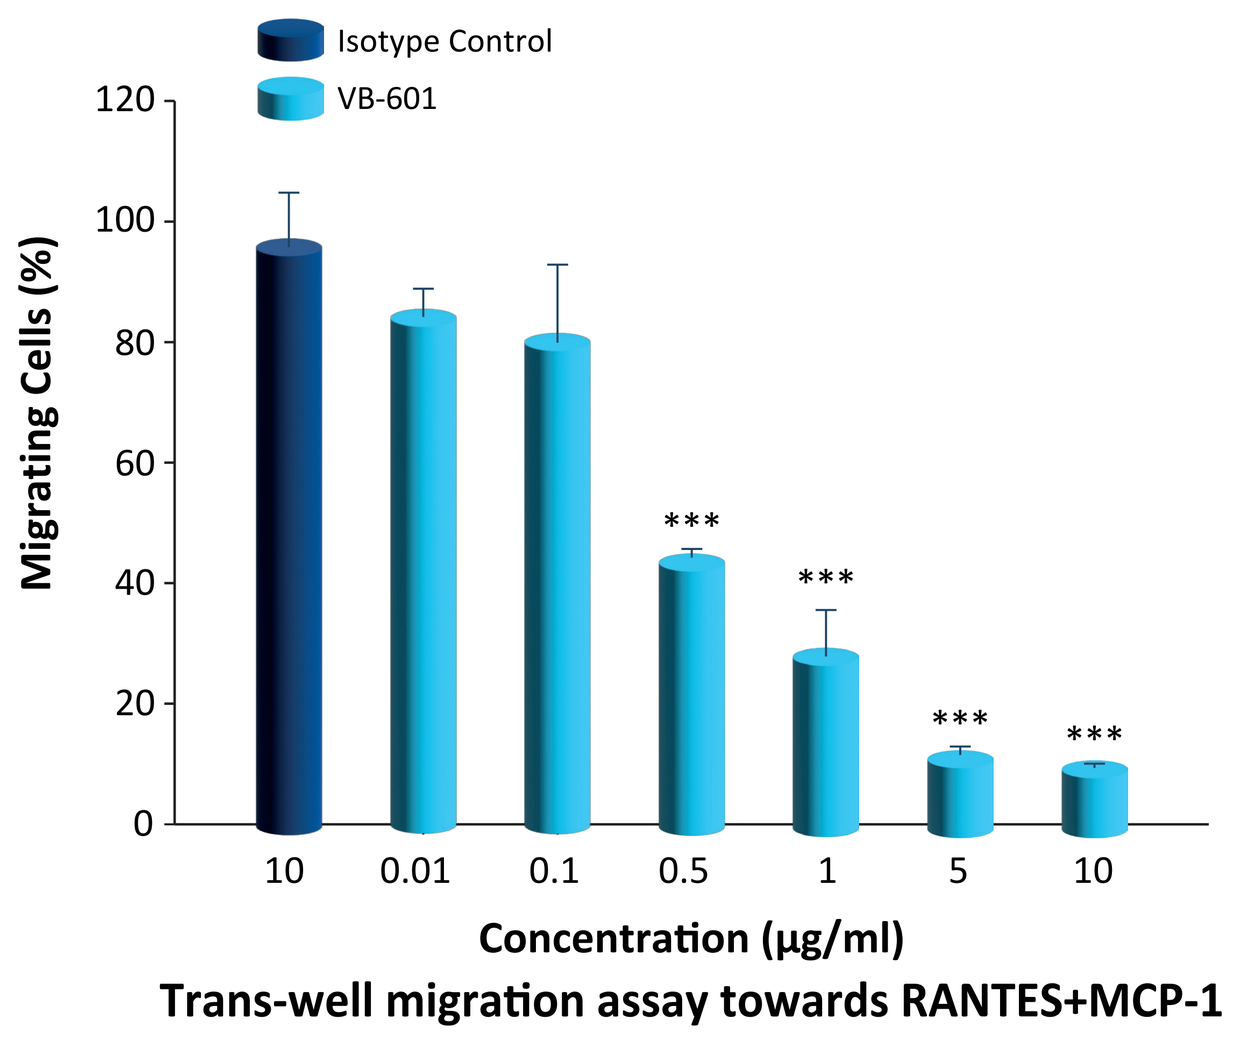
Isotype Control
VB-601
Migrating Cells (%)
120
100
80
60
40
20
0

10
0.01
0.1
0.5
1
5
10
Concentration (µg/ml)
Trans-well migration assay towards RANTES+MCP-1

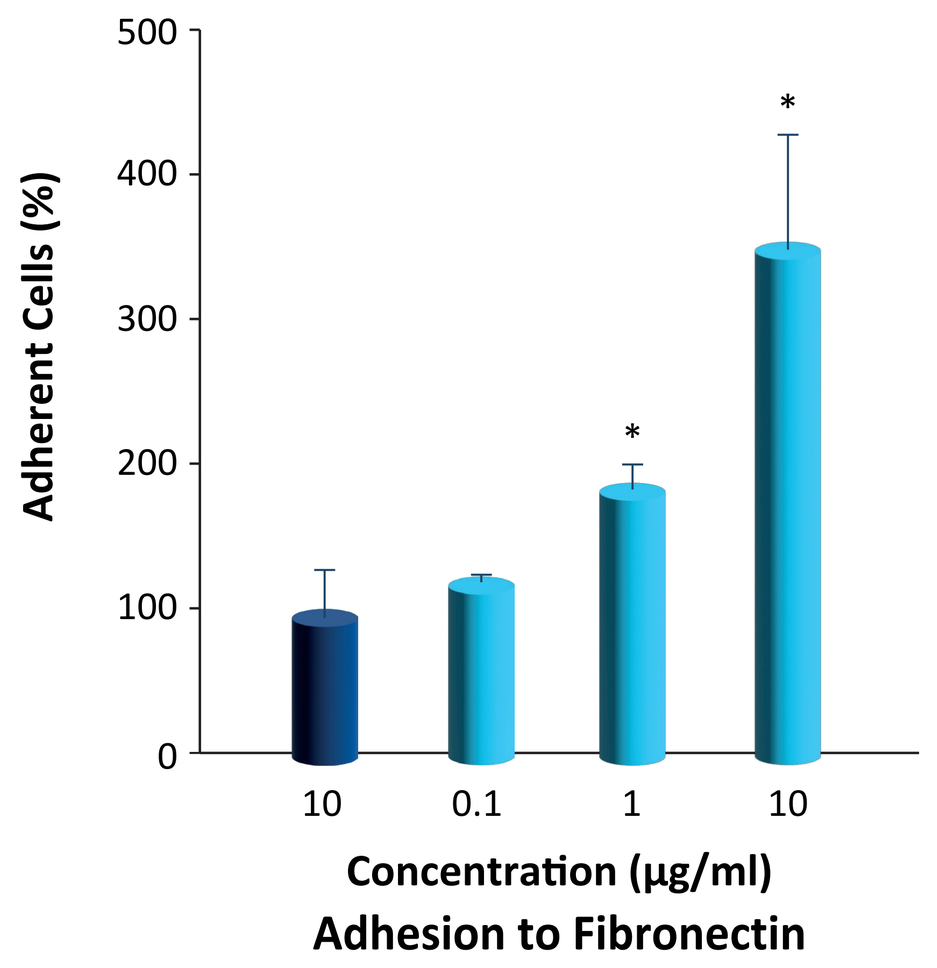
Adherent Cells (%)
500
400
300
200
100
0
*
*
10
0.1
1
10
Concentration (µg/ml)
Adhesion to Fibronectin

VB-601 increases adhesion of human primary monocytes to diverse ECM and adhesion molecules

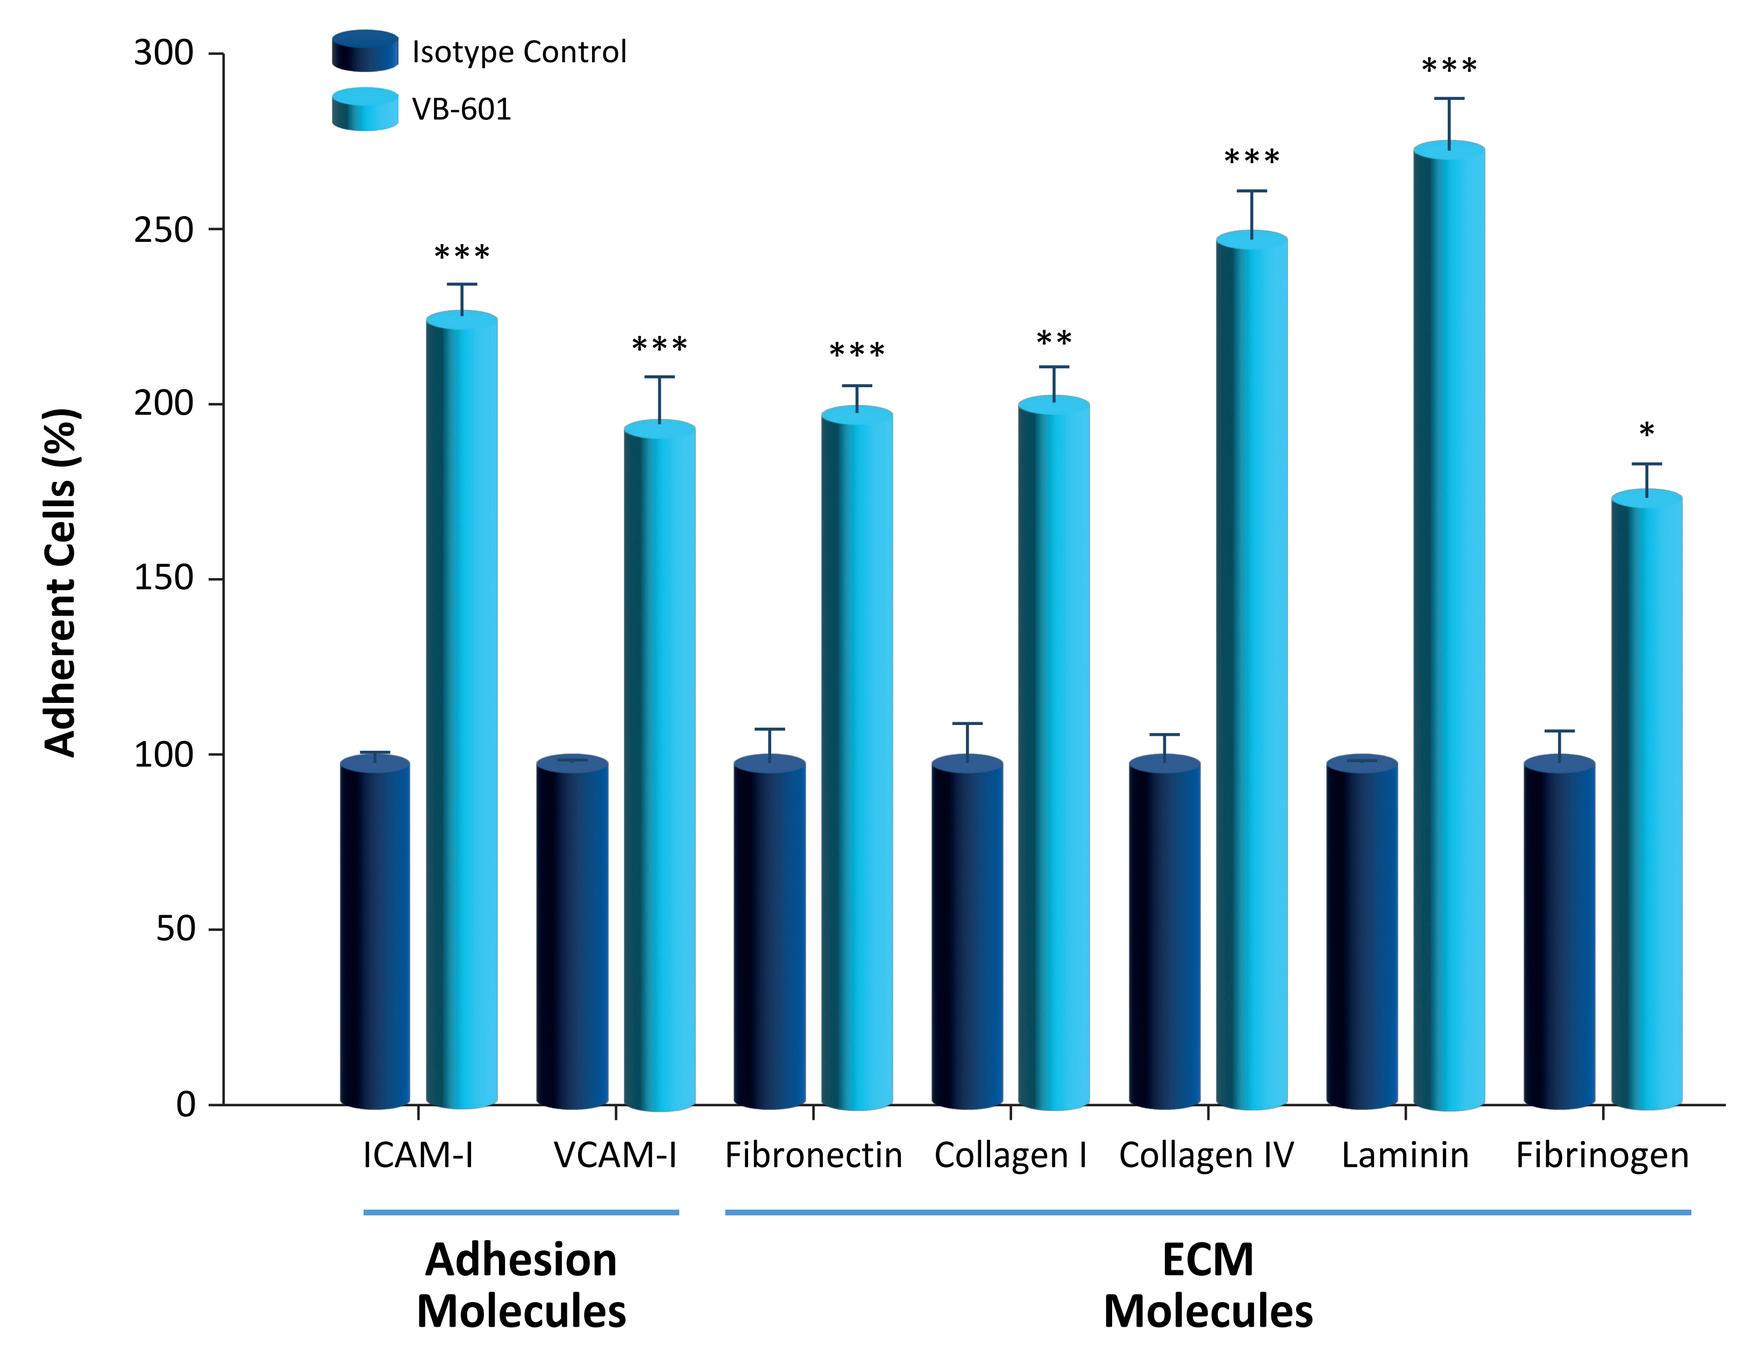
Isotype Control
VB-601
Adherent Cells (%)
300
250
200
150
100
50
0

**

*
ICAM-I
VCAM-I
Fibronectin
Collagen I
Collagen IV
Laminin
Fibrinogen
Adhesion Molecules
ECM Molecules

Silencing or blocking MOSPD2 in monocytes induces αLβ2 integrin transition to high affinity conformation

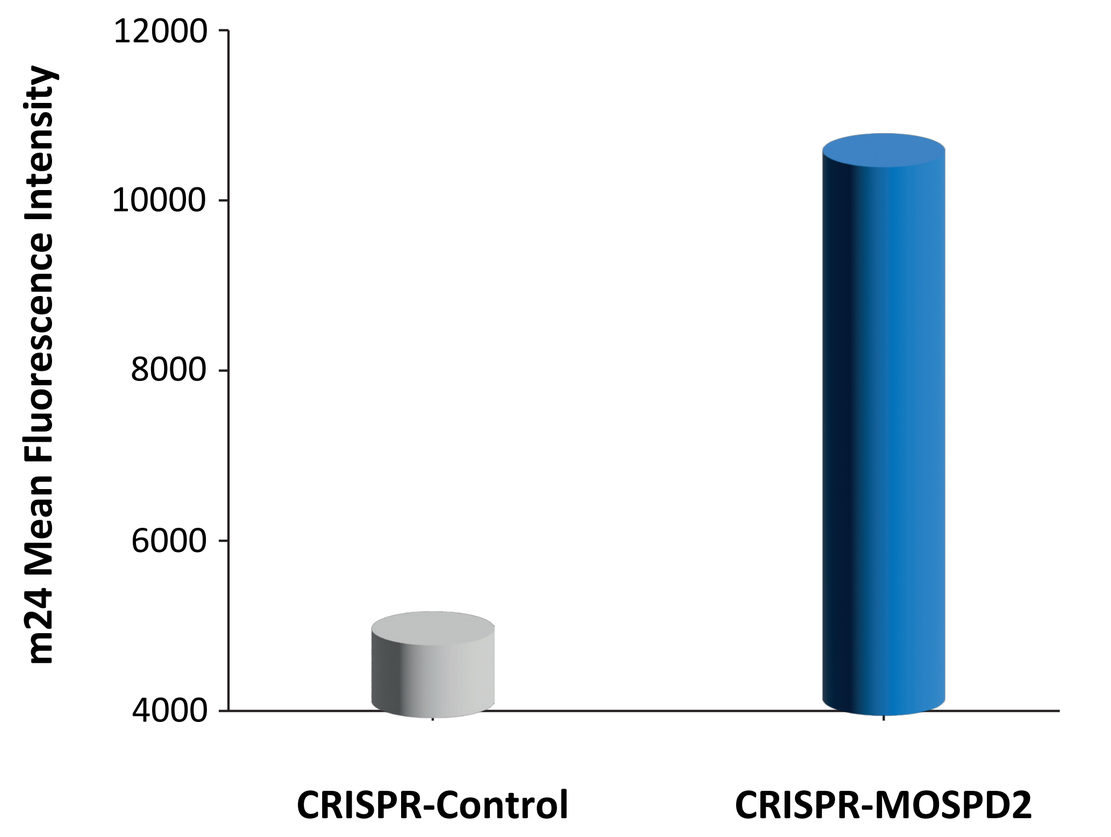
m24 Mean Fluorescence Intensity
12000
10000
8000
6000
4000
CRISPR-Control
CRISPR-MOSPD2

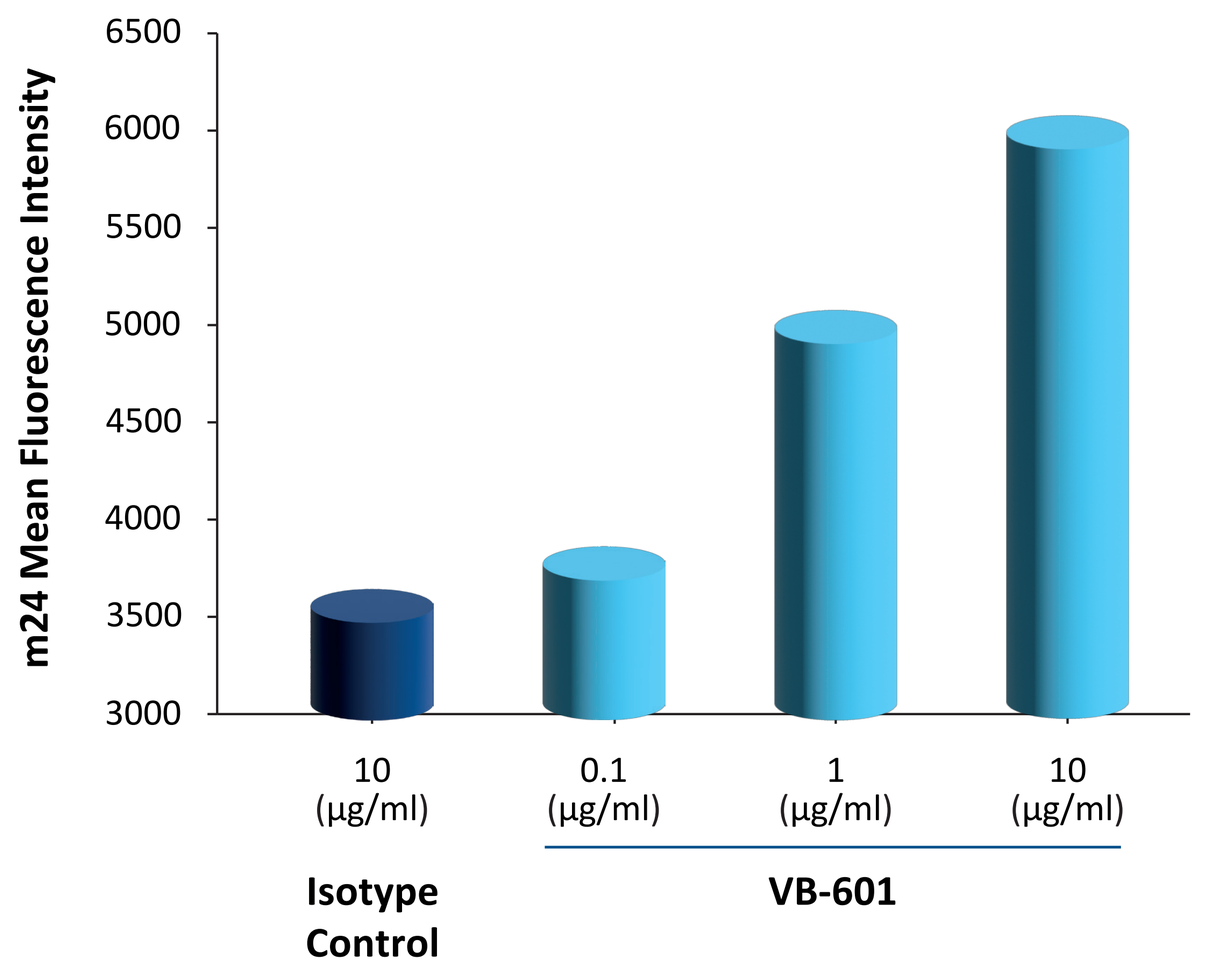
m24 Mean Fluorescence Intensity
6500
6000
5500
5000
4500
4000
3500
3000
10 (µg/ml)
0.1 (µg/ml)
1 (µg/ml)
10 (µg/ml)
Isotype Control
VB-601

Clone m24 binds the extended-open high affinity conformation of αLβ2 (also known as LFA-1) integrin

Results represents high affinity conformation by Flow Cytometry

Co-Immunoprecipitation discovers binding interaction between MOSPD2 and Integrin-β2

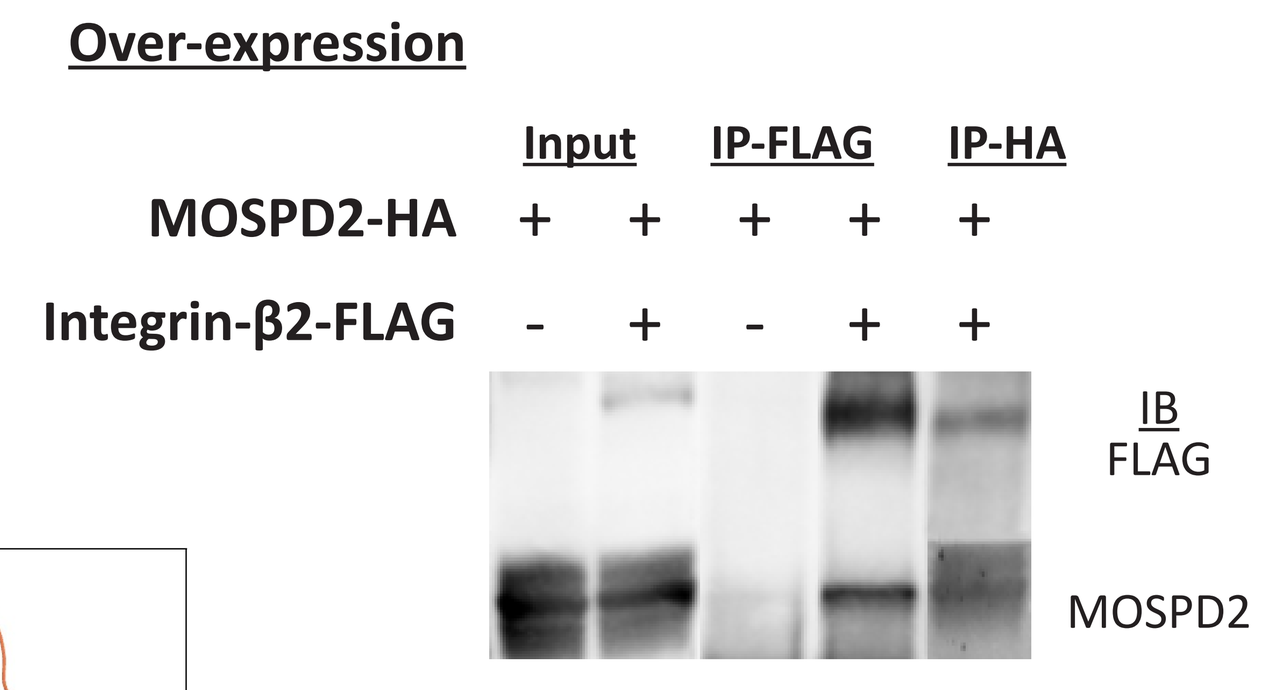
Over-expression
Input
IP-FLAG
IP-HA
MOSPD2-HA + + + + +
Integrin-β2-FLAG - + - + +
IB
FLAG
MOSPD2

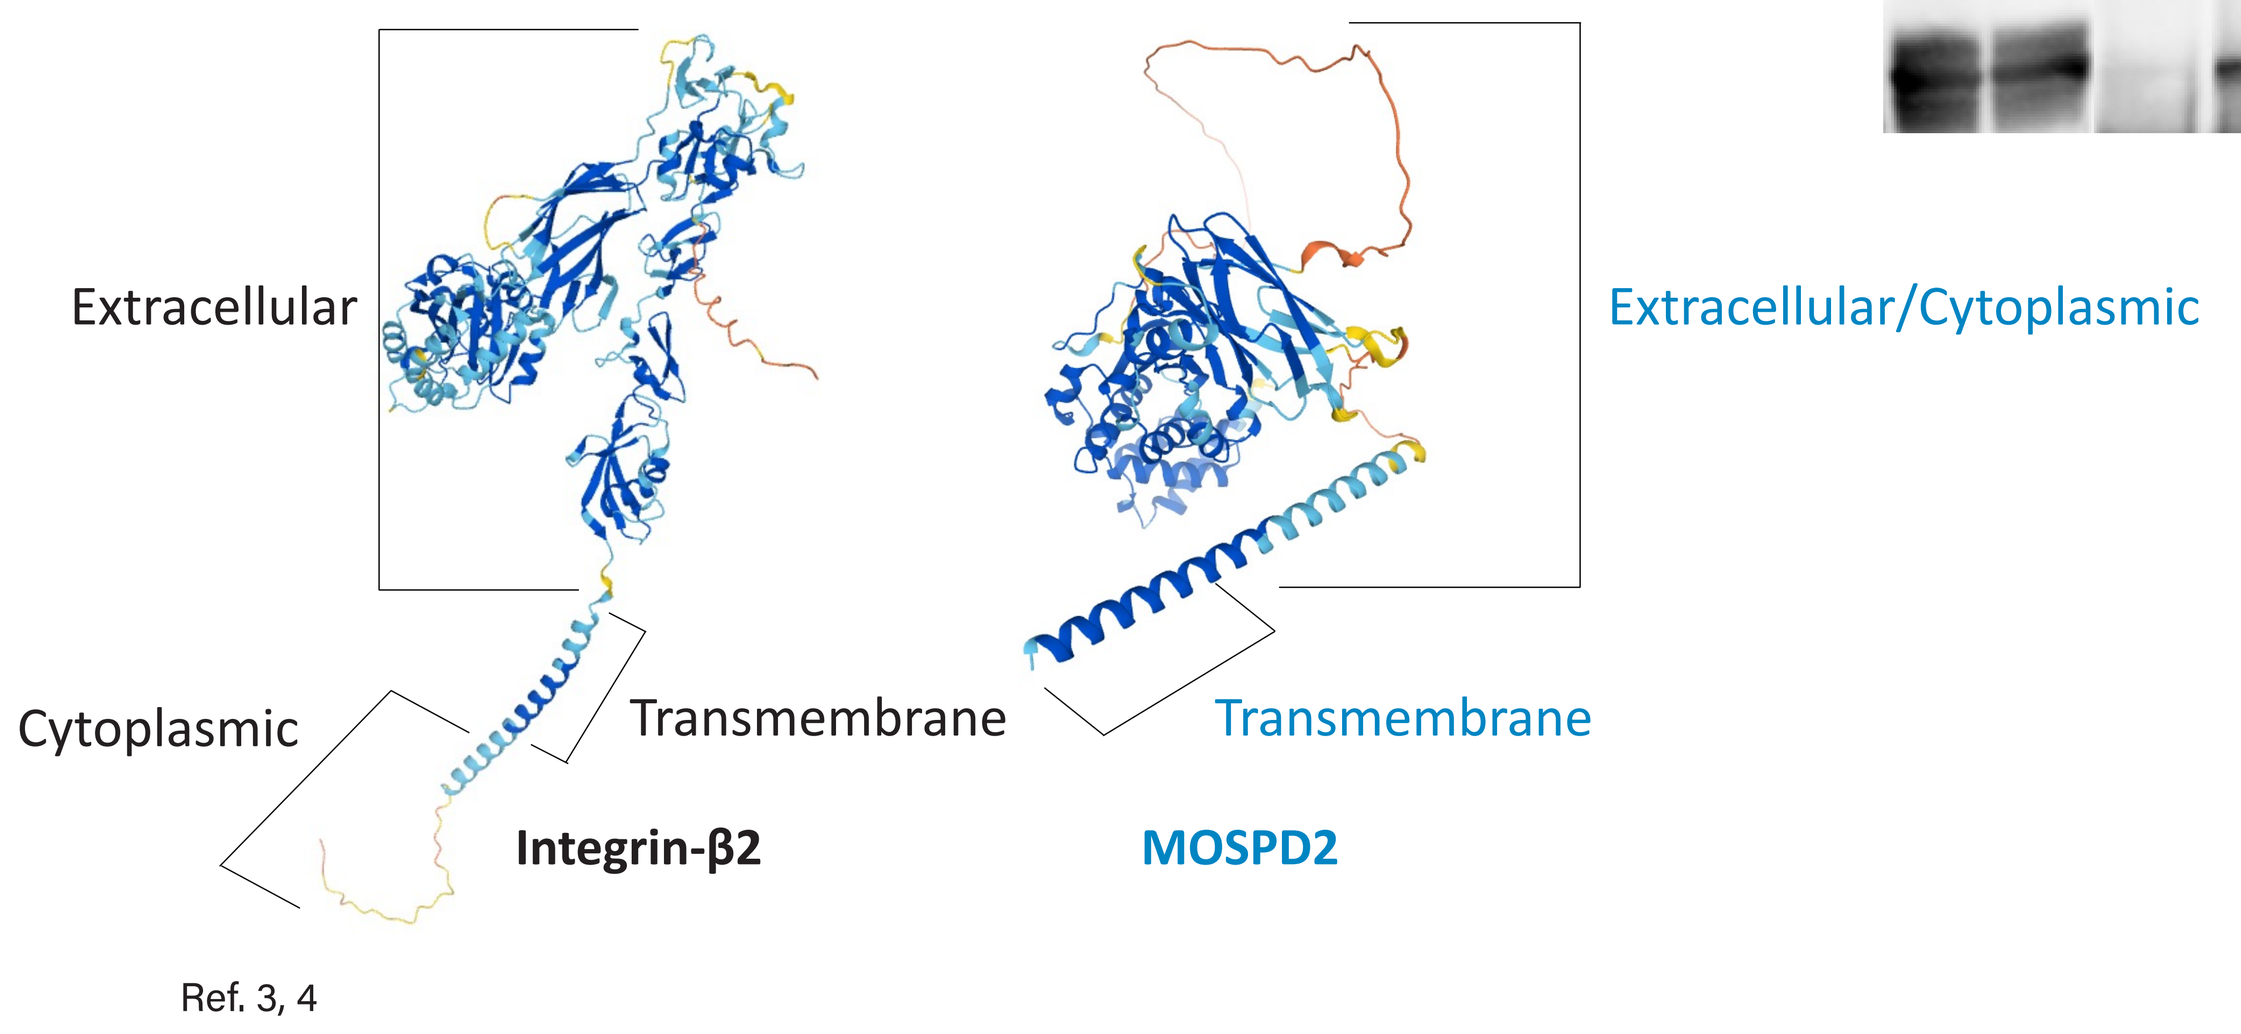
Extracellular
Cytoplasmic
Transmembrane
Integrin-β2
Extracellular/Cytoplasmic
Transmembrane
MOSPD2

Ref. 3, 4

Conclusions

MOSPD2 - "the Monowalk Protein", regulates the activation state of integrin-β2 on monocytes - (A) MOSPD2 enables the monocyte to adhere and migrate (B) Treatment with anti-MOSPD2 mAb - VB-601, blocks monocyte ability to detach from the tissue and migrate.

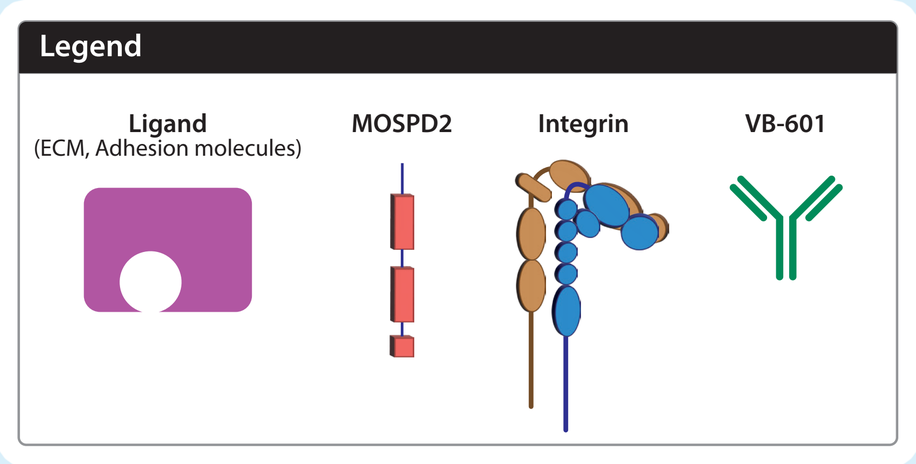
Legend
Ligand (ECM, Adhesion molecules)
MOSPD2
Integrin
VB-601

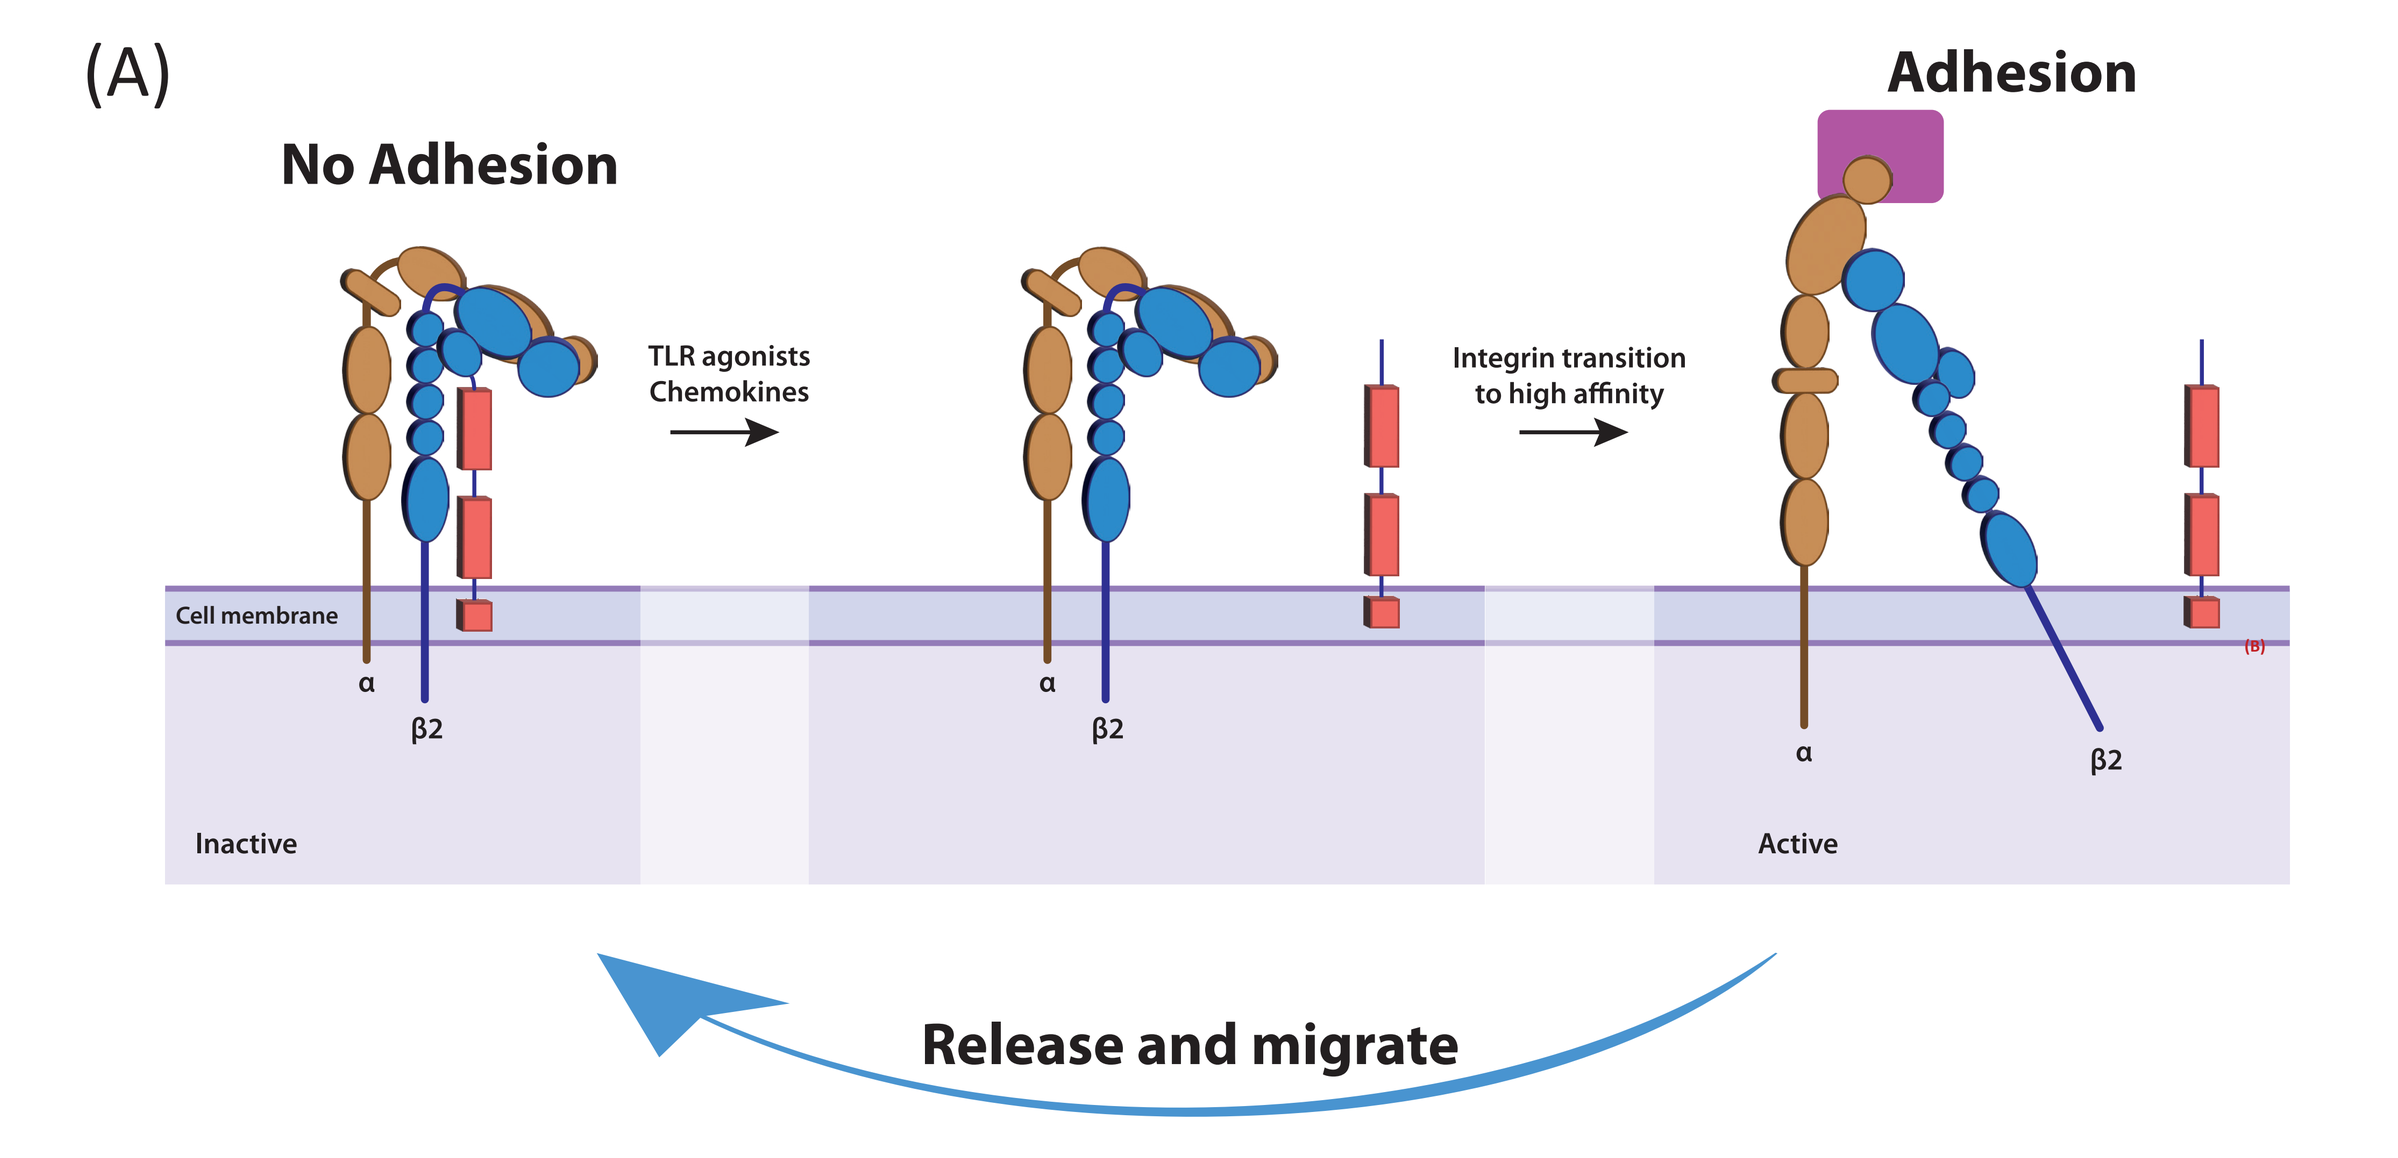
(A)
No Adhesion
Adhesion
TLR agonists Chemokines
Integrin transition to high affinity
Cell membrane
α
β2
Inactive
Active
Release and migrate

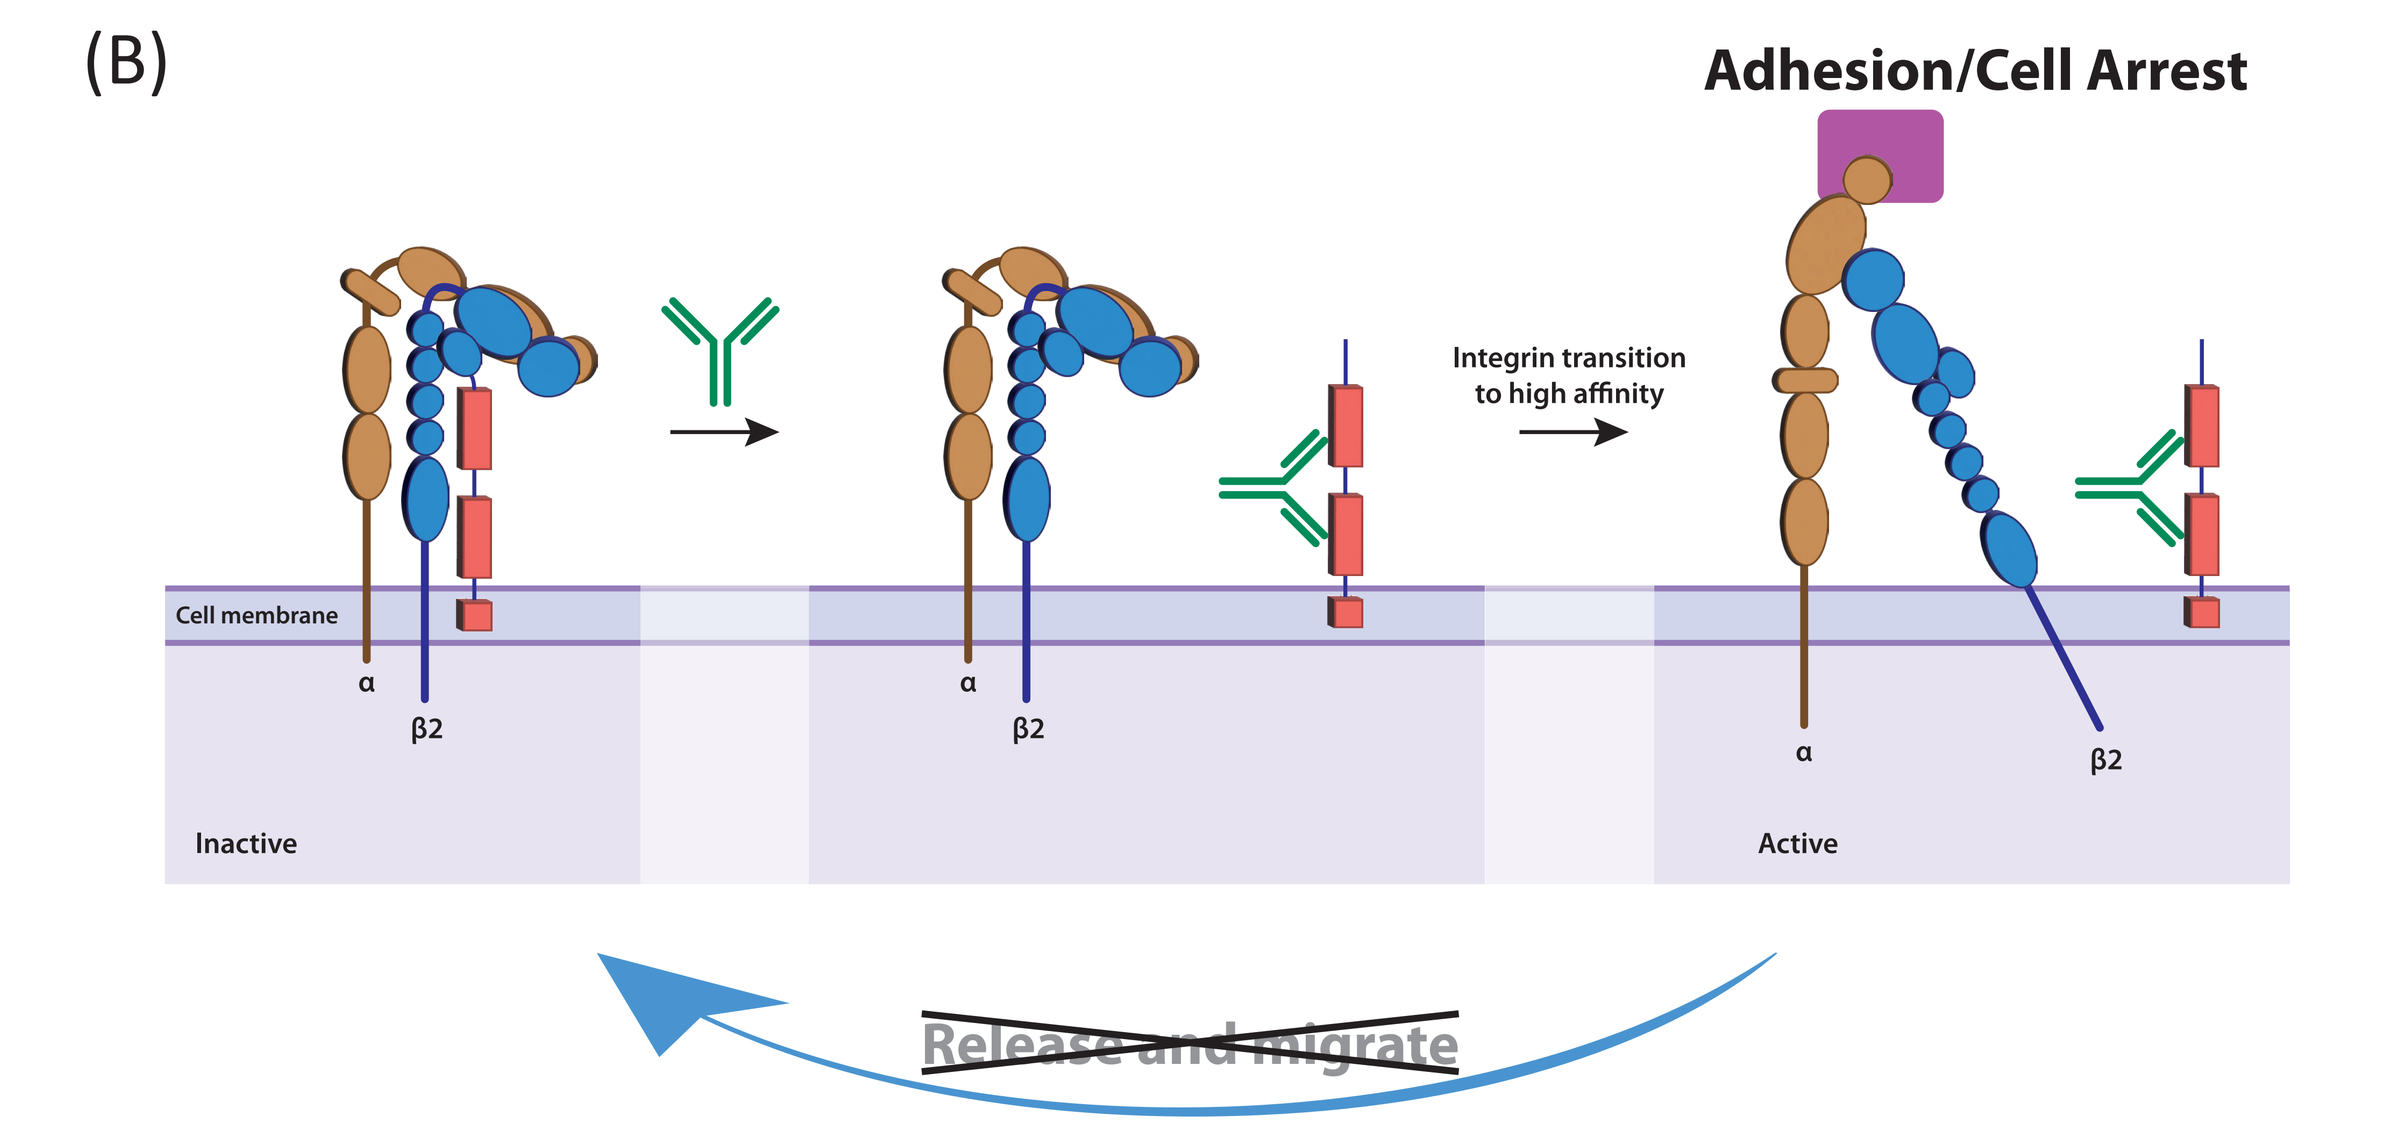
(B)
Adhesion/Cell Arrest
Integrin transition to high affinity
Cell membrane
α
β2
Inactive
Active
~~Release and migrate~~

- MOSPD2 - "the Mono-walk Protein", is a surface protein expressed on myeloid cells
- At the molecular level, MOSPD2 binds integrin β2 to regulate the activation state of αLβ2, thus controlling the dynamic process of adhesion and release from endothelial adhesion molecules and ECM components, which is required for monocyte \`walking\`
- Targeting MOSPD2 via knockout or with the humanized anti-MOSPD2 mAb VB-601 increases adhesion and therefore inhibits migration of monocytes
- Targeting MOSPD2 is a novel and differentiated approach to regulating inflammation:
 - Current agents are mainly directed against lymphocytes, while MOSPD2 regulates monocytes
 - MOSPD2 mAb arrests monocyte from \`walking\` to inflamed tissues through a distinct mechanism of chemokines and their cognate receptors, thereby overcoming the broad redundancy of these pathways
- Proof of concept demonstrated:
 - Promising results in preclinical models of MS, RA, IBD, NASH
 - VB-601 was found to inhibit ex-vivo the migration of monocytes isolated from patients with MS, RA, PsA, Ulcerative Colitis and Crohn's disease
- VBL's lead candidate VB-601 is a humanized IgG1 mAb developed as subcutaneous formulation for treatment of people with chronic immune-inflammatory diseases. VB-601 is being advanced toward first-in-human study, expected 2H 2022

References: 1. Mendel, I. J Immunol. 198: 2125-2132 (2017), 2. Yacov, N. Clin Exp Immunol. 201: 105-120 (2020), 3. Jumper, J. Nature. 596: 583–589 (2021), 4. Varadi, M. Nucleic Acids Research 50: Issue D1 D439–D444 (2021)

www.vblrx.com