Uncovering the unique properties of cell derived, vesicles, a novel carrier with therapeutic promises

Hui-Chong Lau[1], Jihye Lee[1], Dong Woo Han[1], Jinhee Park[1], Soyeon Kim[1] Jae Young Kim[1], Suhee Kim[2], Hye-Jung Kim[2], and Seung Wook Oh[1] *
[1] MDimune Research Institute, MDimune Inc., Seoul, Republic of Korea
[2]New Drug Development Center, KBIO Osong Medical Innovation Foundation, Cheongju-si, Chungbuk, Republic of Korea
*Correspondence: swoh@mdimune.com

INTRODUCTION & EXTRUSION TECHNOLOGY

Extracellular vesicles (EVs), or more frequently known as exosomes, have recently emerged as novel therapeutics in various clinical applications. Despite the growing number of evidences demonstrating that EVs are a crucial mediator of intercellular communication, challenges to commercialize EVs in clinical applications remain largely unresolved. Here, we developed a scalable manufacturing process using extrusion technology to produce cell derived vesicles (CDVs), with superior yield compared to other EVs. Moreover, characterization of CDVs at physical, biochemical and molecular levels provides substantial evidence to further support the therapeutic potentials of CDVs.

(A)

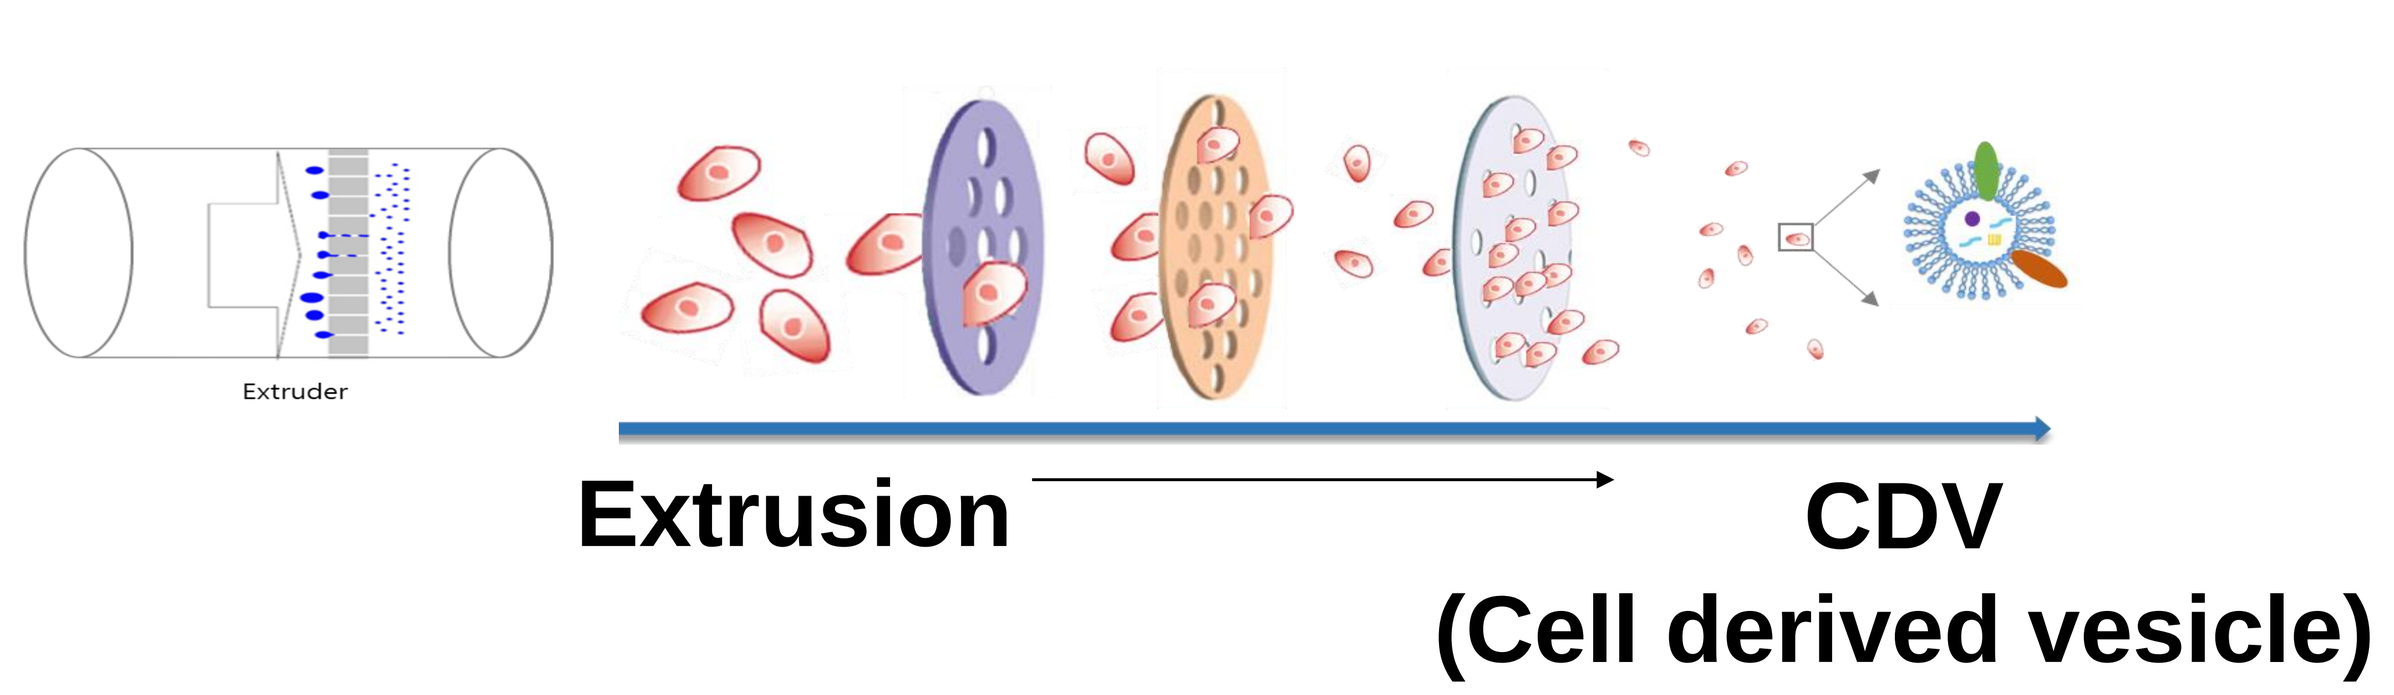
Extruder
Extrusion
CDV
(Cell derived vesicle)

(B)

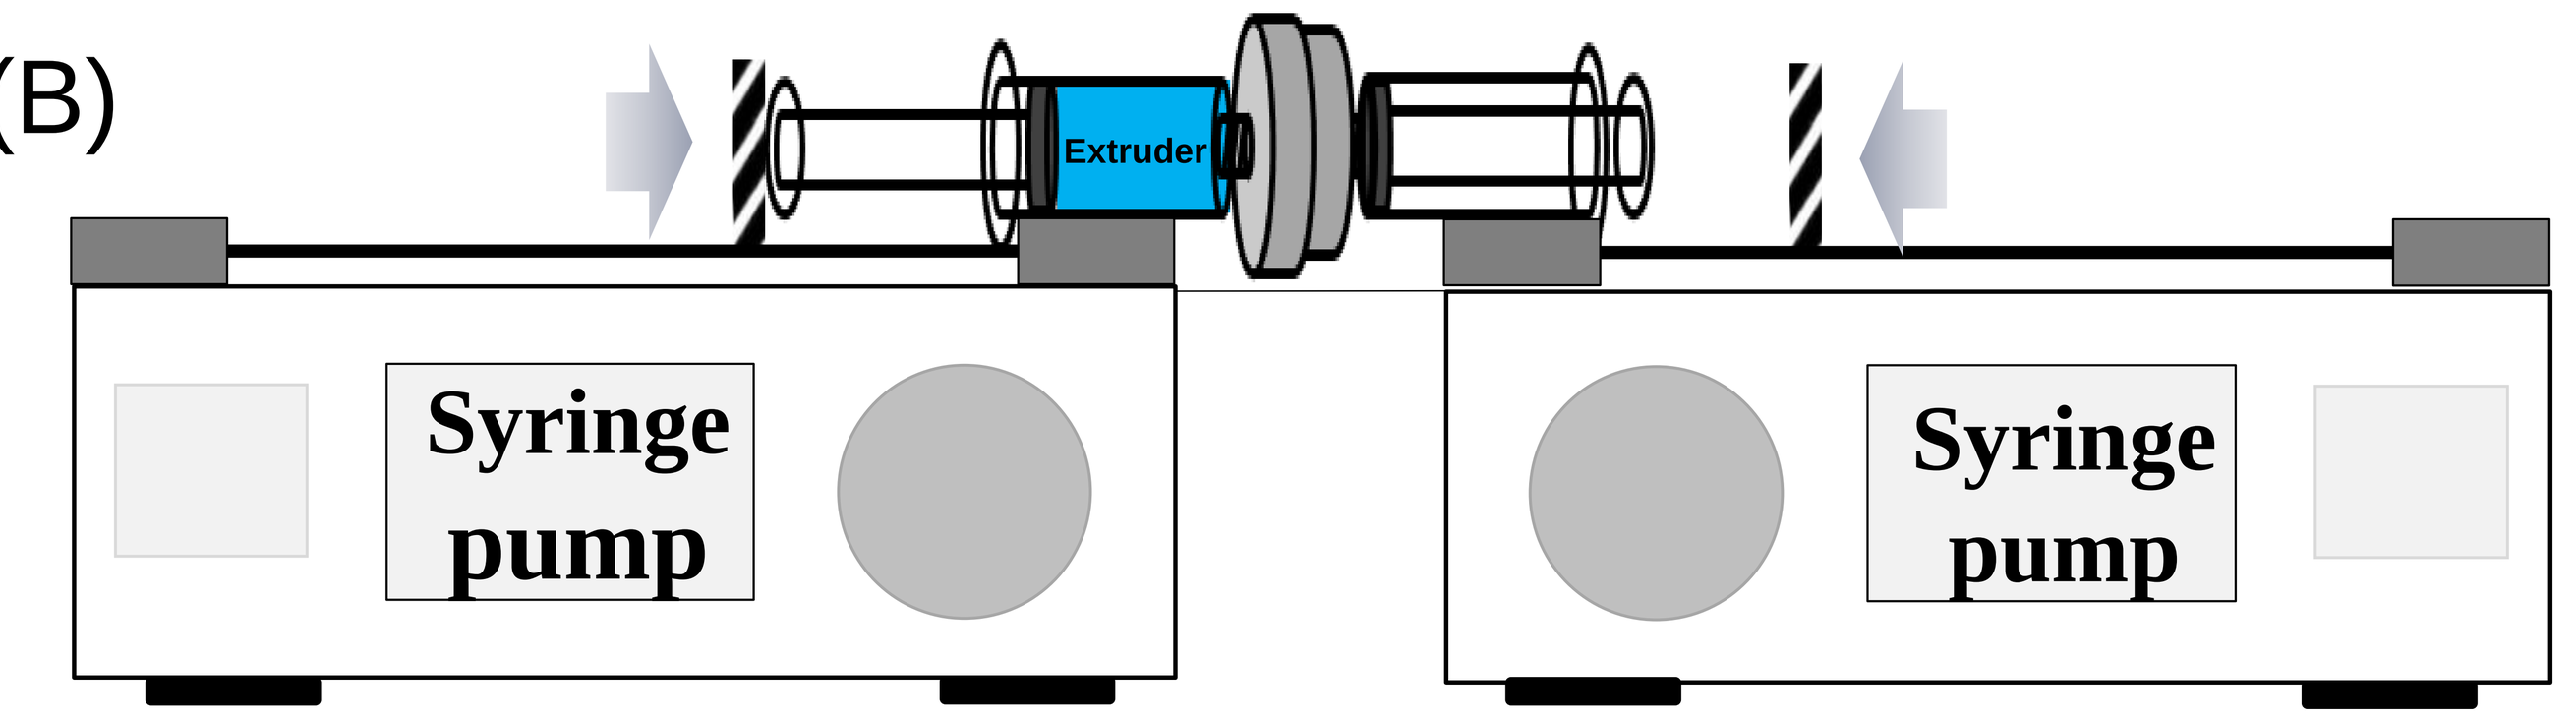
Extruder
Syringe pump
Syringe pump

Figure 1: (A) Manufacturing-scale production of CDVs using serial extrusion with different membrane pore sizes to produce CDVs of the size of interest. (B) The syringe-type extruder developed at MDimune for CDV production.

PRODUCTION PROCESS, CHARACTERIZATION & RELEASE CRITERIA

(A)

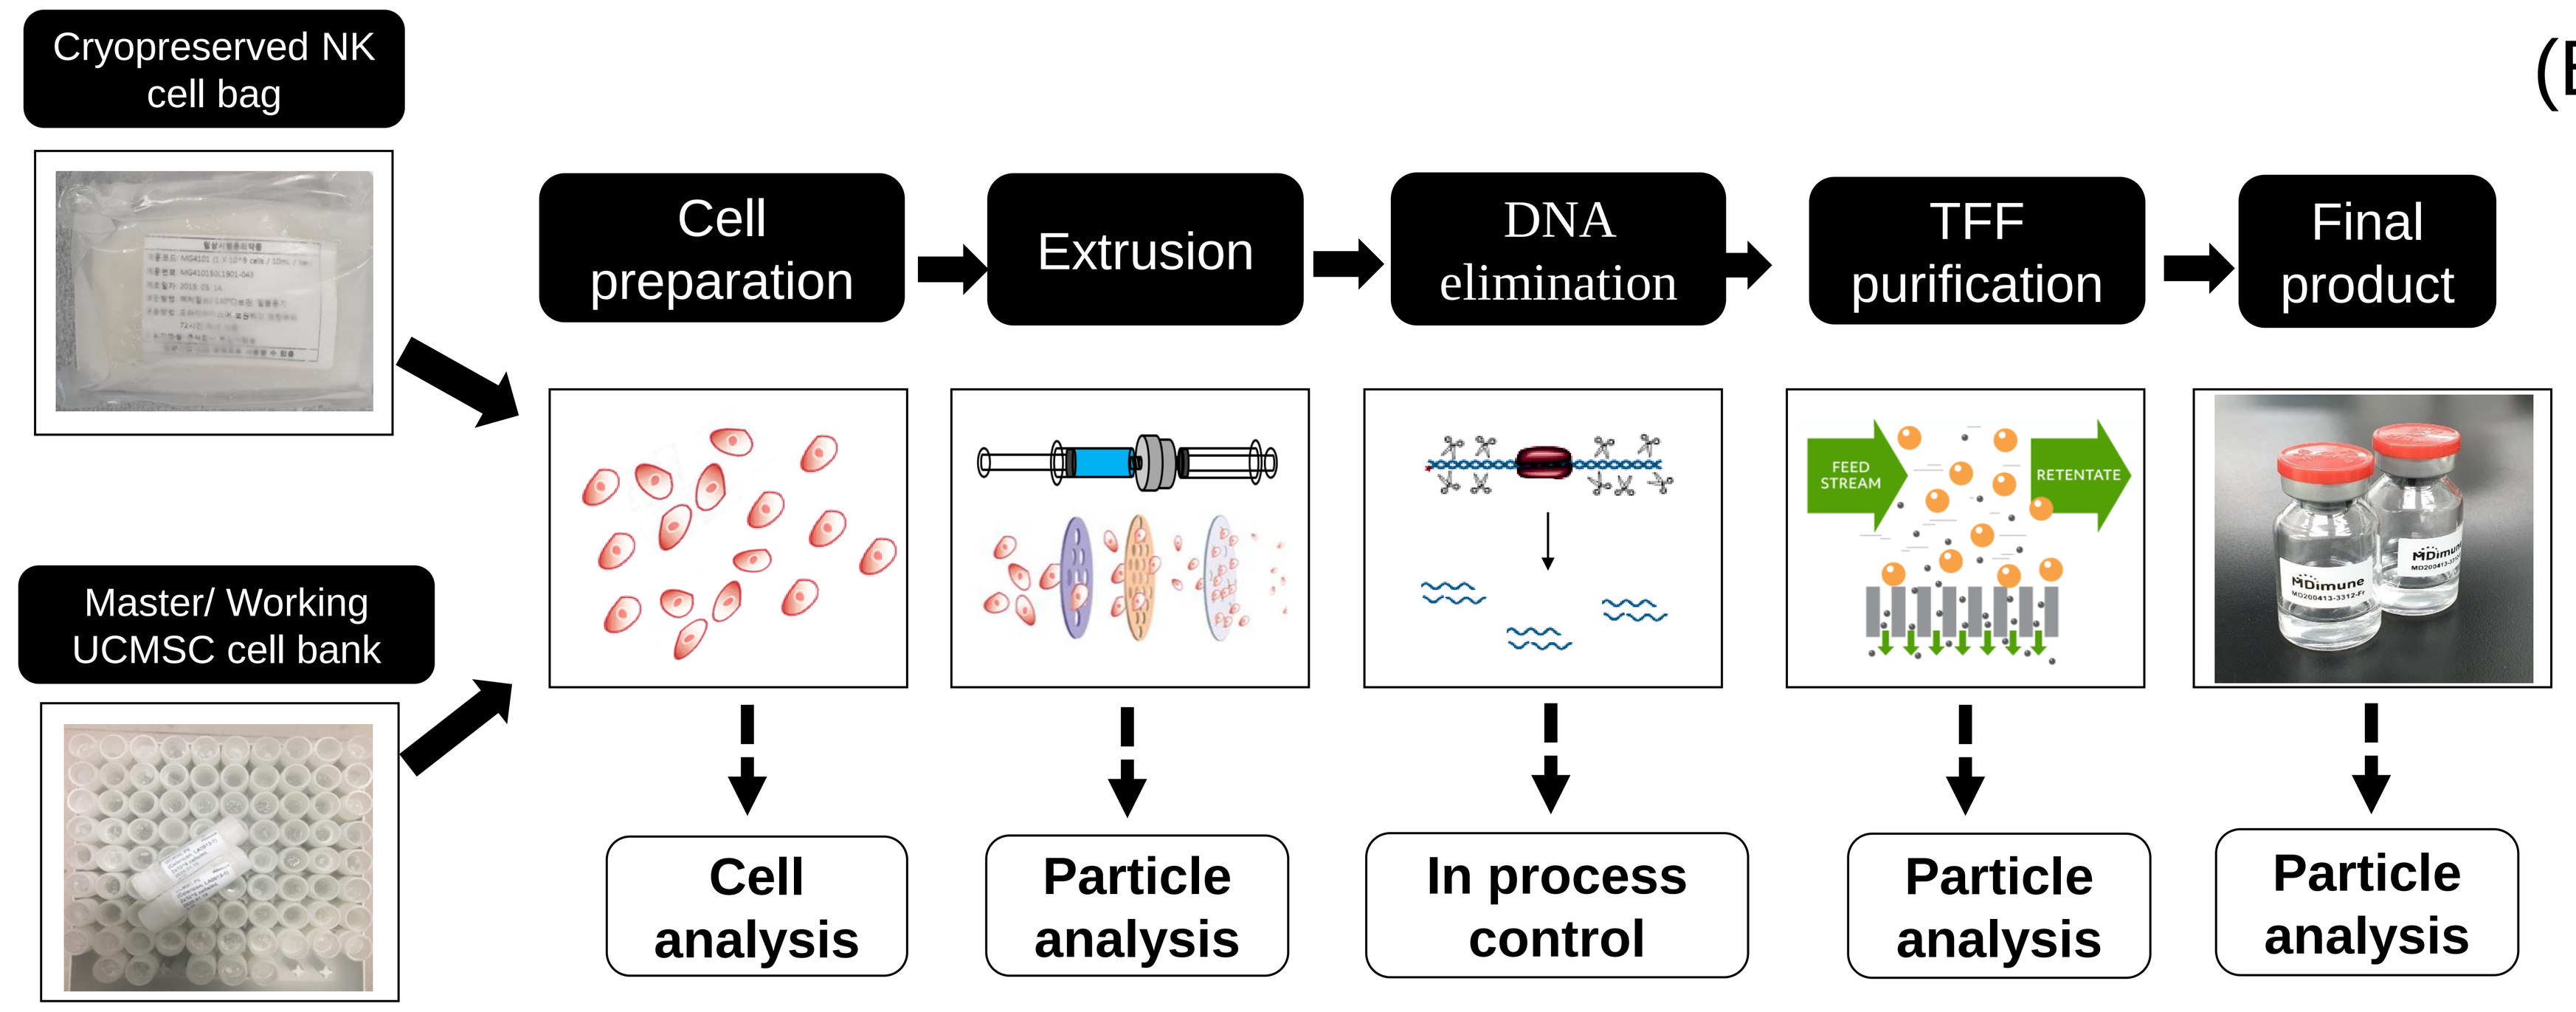
Cryopreserved NK cell bag
Master/ Working UCMSC cell bank
Cell preparation
Extrusion
DNA elimination
TFF purification
Final product
FEED STREAM
RETENTATE
Cell analysis
Particle analysis
In process control
Particle analysis
Particle analysis

(B)

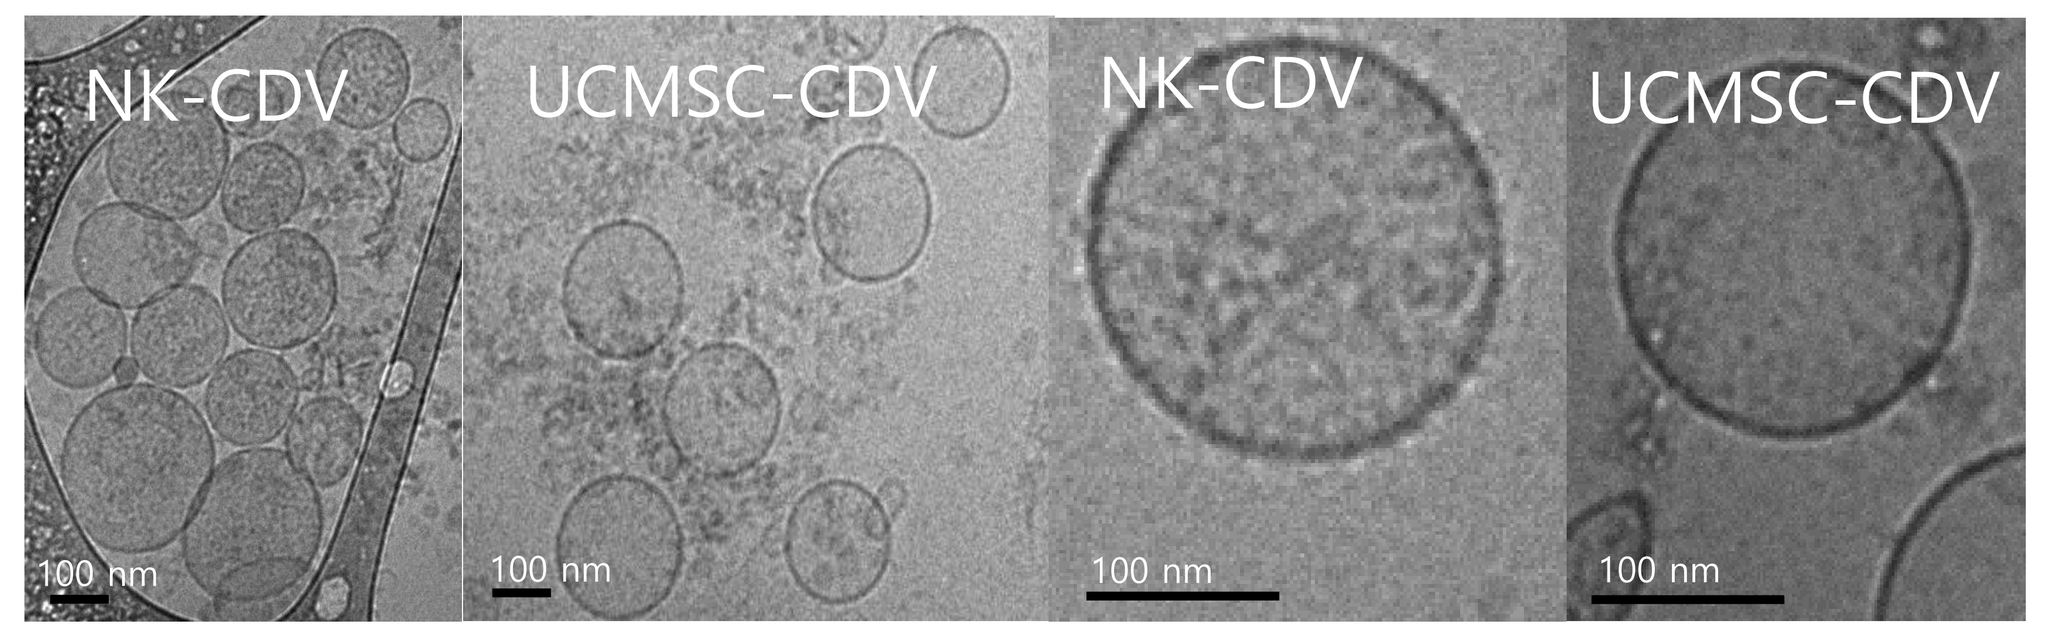
NK-CDV
UCMSC-CDV
NK-CDV
UCMSC-CDV
100 nm
100 nm
100 nm
100 nm

Figure 2: (A) A process to produce CDVs from different cell sources and QC steps to control the quality of CDVs. (B) Representative images of CDVs examined using cryo-TEM. CDV has a round and spherical shape with diameter ranges from 50-250 nm. Lipid layer of CDV can be clearly observed under microscope.

Table 1: QC parameter and current release criteria of CDVs

Category	Parameter	Instrument/method	Current release criteria	
			NK-CDV	UCMSC-CDV
Purified CDV	Size (nm)	DLS	150~200 nm	80-150
	PDI	DLS	0.1~0.3	0.2-0.5
	Particles/cell	NTA	2.7E+3	4.4E+4
	Extracted DNA (ng/1E+12 particles)	Pico green dsDNA assay kit	51.3	96.6
	Benzonase (Unit/1E+12 particles)	ELISA	≤20	<38
	Surface marker	FACS (Aldehyde sulfate bead)	>70% CD11a, CD56	>70% CD63, CD73, CD90, CD29, CD44

Physical and biochemical properties of CDV were characterized to control the quality of CDV at each batch of production. Current release criteria was established based on the results from multiple batches of production.

MULTI-OMICS ANAYSIS

Multi-omics profiling was carried out with three independent batches of NK cells and NK-CDV to comprehend the fundamental understanding of the molecular contents of CDVs. In proteomics analysis, to meet the criteria of significance, all genes with fold change > 2 and p-value < 0.05 were used in this study.

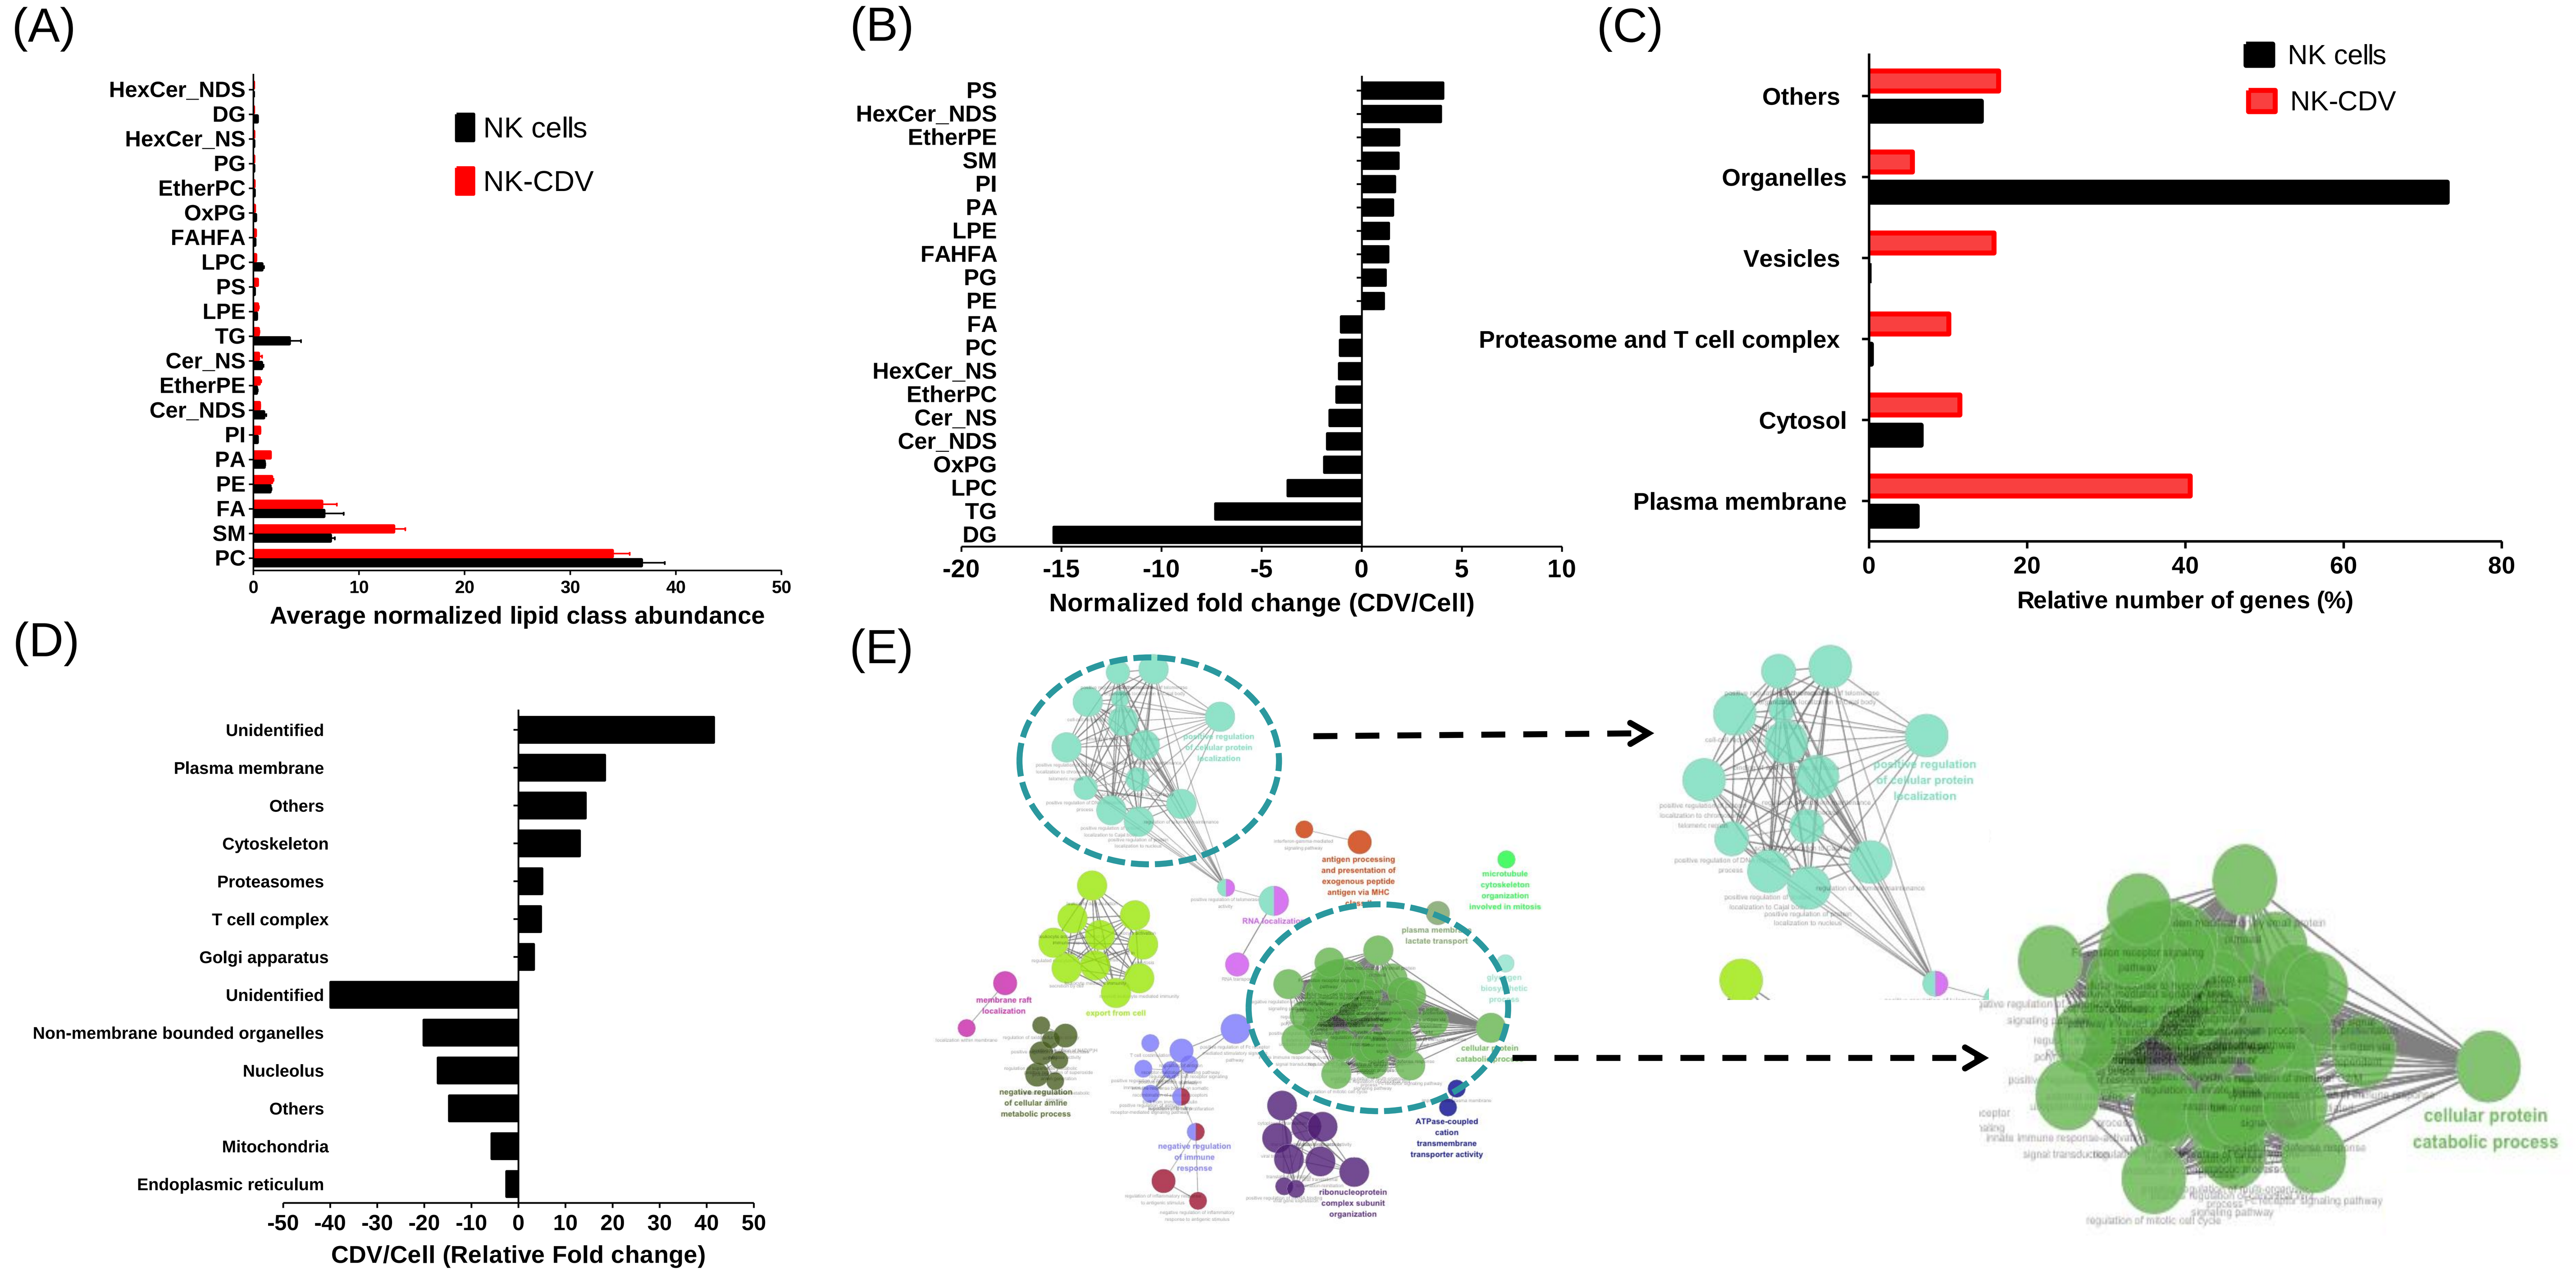
(A)
HexCer_NDS
DG
HexCer_NS
PG
EtherPC
OxPG
FAHFA
LPC
PS
LPE
TG
Cer_NS
EtherPE
Cer_NDS
PI
PA
PE
FA
SM
PC
NK cells
NK-CDV
0 10 20 30 40 50
Average normalized lipid class abundance
(B)
PS
HexCer_NDS
EtherPE
SM
PI
PA
LPE
FAHFA
PG
PE
FA
PC
HexCer_NS
EtherPC
Cer_NS
Cer_NDS
OxPG
LPC
TG
DG
-20 -15 -10 -5 0 5 10
Normalized fold change (CDV/Cell)
(C)
NK cells
NK-CDV
Others
Organelles
Vesicles
Proteasome and T cell complex
Cytosol
Plasma membrane
0 20 40 60 80
Relative number of genes (%)
(D)
Unidentified
Plasma membrane
Others
Cytoskeleton
Proteasomes
T cell complex
Golgi apparatus
Unidentified
Non-membrane bounded organelles
Nucleolus
Others
Mitochondria
Endoplasmic reticulum
-50 -40 -30 -20 -10 0 10 20 30 40 50
CDV/Cell (Relative Fold change)
(E)
positive regulation of cellular protein localization
antigen processing and presentation of exogenous peptide antigen via MHC class I
microtubule cytoskeleton organization involved in mitosis
plasma membrane lactate transport
RNA localization
export from cell
membrane raft localization
negative regulation of cellular amine metabolic process
negative regulation of immune response
ATPase-coupled cation transmembrane transporter activity
ribonucleoprotein complex subunit organization
cellular protein catabolic process

Figure 3: Multi-omics analysis of NK cells and NK-CDVs. (A) Distribution of lipid class in NK cells and NK-CDVs based on average abundances. (B) Enrichment of lipid class based on normalized fold change of lipid (CDV/cell). (C) Gene Ontology (GO) analysis of list of genes identified from NK cells and NK-CDV. The cellular components were further grouped into 6 main categories. The main cellular component of CDVs is derived from plasma membrane. This result was coherent with the western blot study (data not shown), in which plasma membrane is enriched in NK-CDV. (D) Enriched cellular component in CDVs based on representative GO cellular component. (E) Protein-protein interaction network of NK-CDVs was constructed using Cytoscape ClueGO. The main clusters of NK-CDVs are associated with cellular protein catabolic process and regulation of protein localization.

PRODUCTION YIELD

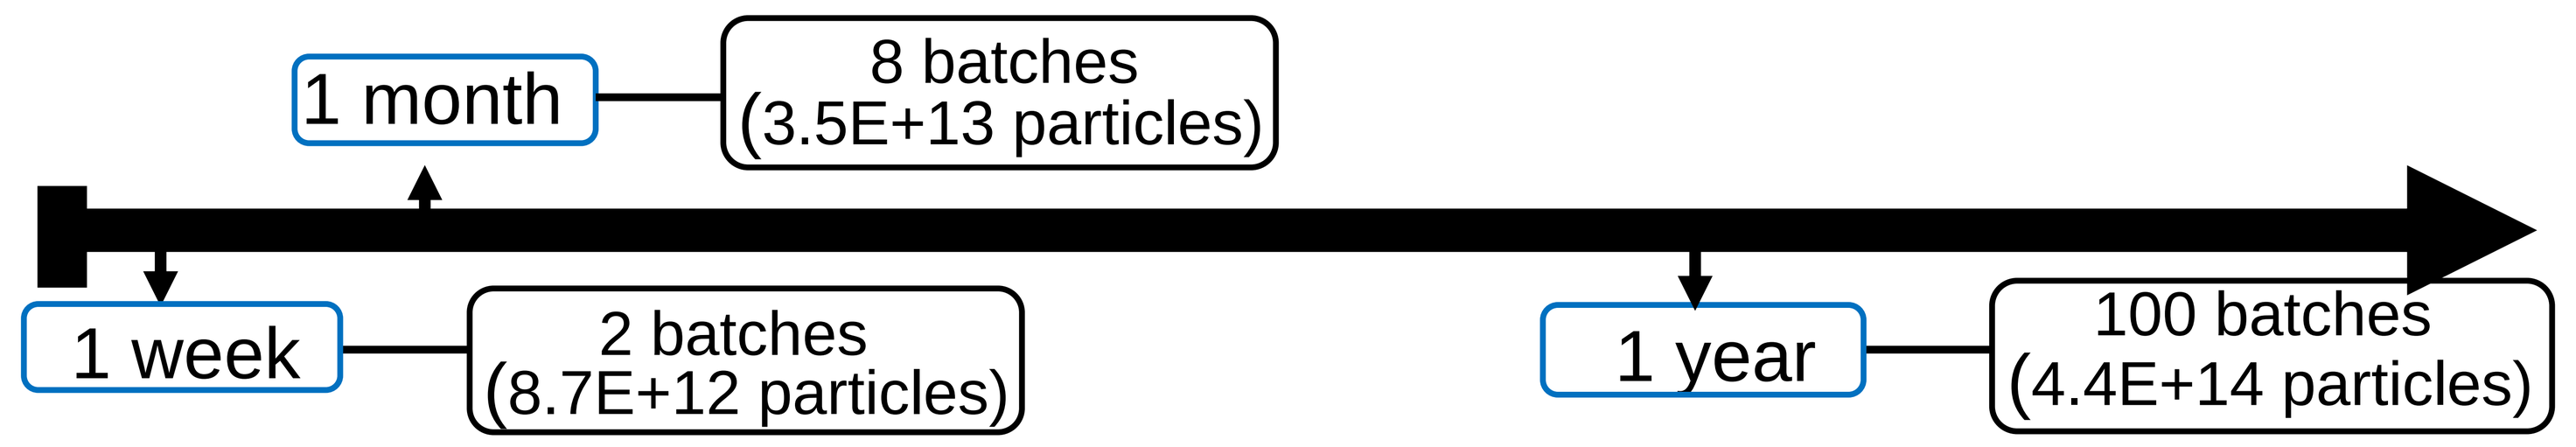
1 month
8 batches (3.5E+13 particles)
1 week
2 batches (8.7E+12 particles)
1 year
100 batches (4.4E+14 particles)

Figure 4: Current production capacity and yield of CDV on weekly, monthly and annual basis. With existing setting, more than 4E+14 particles can be produced for pre-clinical usage. Such setting can be further upgraded to meet the clinical requirement.

CONCLUSIONS

- We developed a scalable manufacturing process and quality control criteria for CDVs.
- Characterization of CDVs reveals physical and biochemical properties shared with other EVs, supporting the therapeutic potentials of CDVs.
- Molecular and biochemical analyses show that the main cellular component of CDV is derived from plasma membrane whereas organelles and non-membrane bound organelles are enriched in cells.
- The current output can support various studies at the preclinical and early clinical stages.